Amatoxin-based antibody-drug conjugates induce immunogenic cell death and improve the anti-tumor efficacy of immune checkpoint inhibitors in humanized mouse models

#1754

AACR Annual Meeting 2022

Christian Orlik, Franziska Ebeling, Kristin Decker, Irina Dranova, Anikó Pálfi, Christoph Müller, Torsten Hechler, Andreas Pahl, and Michael Kulke
Heidelberg Pharma Research GmbH, Gregor-Mendel Straße 22, 68526 Ladenburg, Germany

Heidelberg PHARMA
Focused Cancer Therapies

Introduction

Anti-tumor activity of therapeutic antibodies can be significantly enhanced by conjugation to cytotoxic small molecules. By using such antibody-drug conjugates (ADCs) the toxin is exclusively delivered to target cells and thereby kills only those cells. Beside the currently approved ADCs including Enhertu (HER2), Trodelvy (Trop-2), Blenrep (BCMA), and Zynlonta (CD19), 189 ADCs have entered clinical trials, promising to strengthen the therapeutic capabilities for cancer treatment. Most ADCs are based on few cytotoxic mechanisms with topoisomerase-, microtubule- or DNA-targeting toxins as payloads. Accordingly, the use of new drugs that function via alternative toxicity mechanisms could enhance the therapeutic potential of ADCs. Heidelberg Pharma focuses on **amanitin-based ADCs**, so called **ATAC®**, comprising a new class of ADCs with amanitin as toxic payload (1). Amanitin is the well-known toxin of the amatoxin family which specifically binds to the eukaryotic RNA polymerase II thereby inhibiting the cellular transcription process (2, 3). Heidelberg Pharma also pursues the strategy of **site-specific conjugation** to limit heterogeneity of drug-antibody species, to improve conjugate stability, and to increase the therapeutic window of ADCs. For this purpose, antibodies are engineered at specific locations to incorporate reactive cysteines.

In the current study, we show that ATAC®-based ADCs belong to the class of immune activating drugs which exhibit synergistic anti-tumor efficacy with immune checkpoint inhibitors (ICIs) *in vivo* and induce immunogenic cell death (ICD). ICIs are a class of cancer therapeutics utilizing patients\` immune system to kill cancer cells. ICIs rely on the activity of the immune system to develop their full potential. Therefore, drugs that are heating up cold tumors and make them visible to the patients' immune system and by that enhancing the anti-tumor efficacy of ICIs, e.g., ATAC®, are on high demand for the treatment of tumor patients.

Methods

Cell lines: JIMT-1 and Raji cells were obtained from DSMZ and NCI-N87 from ATCC.

Synthesis of conjugates: Cysteine-reactive amanitin-linker constructs were synthesized at Heidelberg Pharma and were site-specifically conjugated to anti-HER2 (variable domains of trastuzumab, cysteine engineered monoclonal IgG backbone, HDP) and chimeric anti-CD19 antibody (DKFZ Heidelberg, Germany; cysteine engineered monoclonal IgG backbone, HDP). Drug-antibody ratio for amanitin conjugates according to LC-MS analysis was ~2.0 amanitins per IgG.

Flow cytometry: Apoptosis and ICD was determined by staining the cells with Annexin V-FITC, 7AAD and anti-calreticulin-FITC, respectively. Staining was measured by using FACS Lyric (BD Biosciences).

Immunohistochemistry: Tumors were fixed with 4% formalin, embedded in paraffine and stained with the anti-HER2 antibody Trastuzumab (Proteinase K antigen retrieval) followed by an anti-human IgG-HRP antibody.

Immunofluorescence: Tumors were snap frozen and embedded in OCT. 7µm sections were fixed with 4% formalin and stained with anti-human F(ab')2 fragment-AF488, anti-HMGB1-AF647, anti-calreticulin and anti-mouse IgG-AF488.

Animal models: $5x10^6$ JIMT-1 or NCI-N87 cells were injected s.c. into 6-8 weeks old female NMRI Nude mice. When tumor volumes reached a mean of app. 150 mm^3, mice were treated with a single i.v. dose of an anti-HER2 ATAC® (T-Ama). 2-4 weeks after complete tumor remission (CR) was achieved, mice were re-inoculated with $5x10^6$ tumor cells.

HER2 (1+) TNBC PDX studies were performed at XenTech (Evry, France). Mice were treated with a single dose of anti-HER2 ATAC® i.v. at a mean tumor volume of 170 mm^3.

$2.5x10^6$ Raji cells in a mix with $1x10^7$ human PBMCs were injected s.c. into 6-8 weeks old NOD Scid mice. On the same day, treatment was initiated with either a single i.v. dose of anti-CD19 ATAC, Ipilimumab (q3dx5), Pembrolizumab (q3dx5), Avelumab (q3dx6) or a combination of ATAC®-based ADC and an ICI.

Results

1. ATAC®-treatment of homogenous cell line-derived xenografts leads to complete tumor remission and immunity towards tumor re-challenge.

HER2 expressing homogenous CDX model of breast (JIMT-1) or gastric carcinoma (NCI-N87) in NMRI nude mice were treated with a single i.v. dose of anti-HER2 ATAC® (T-Ama). ATAC®-treatment led to a complete tumor remission (CR) as compared to control treatment (Figure 1).

Interestingly, mice re-challenged with the same tumor cell line after CR achieved by ATAC®-treatment, developed tumors with significantly lower tumor volume than mice in which the tumor was inoculated for the first time (Figure 2).

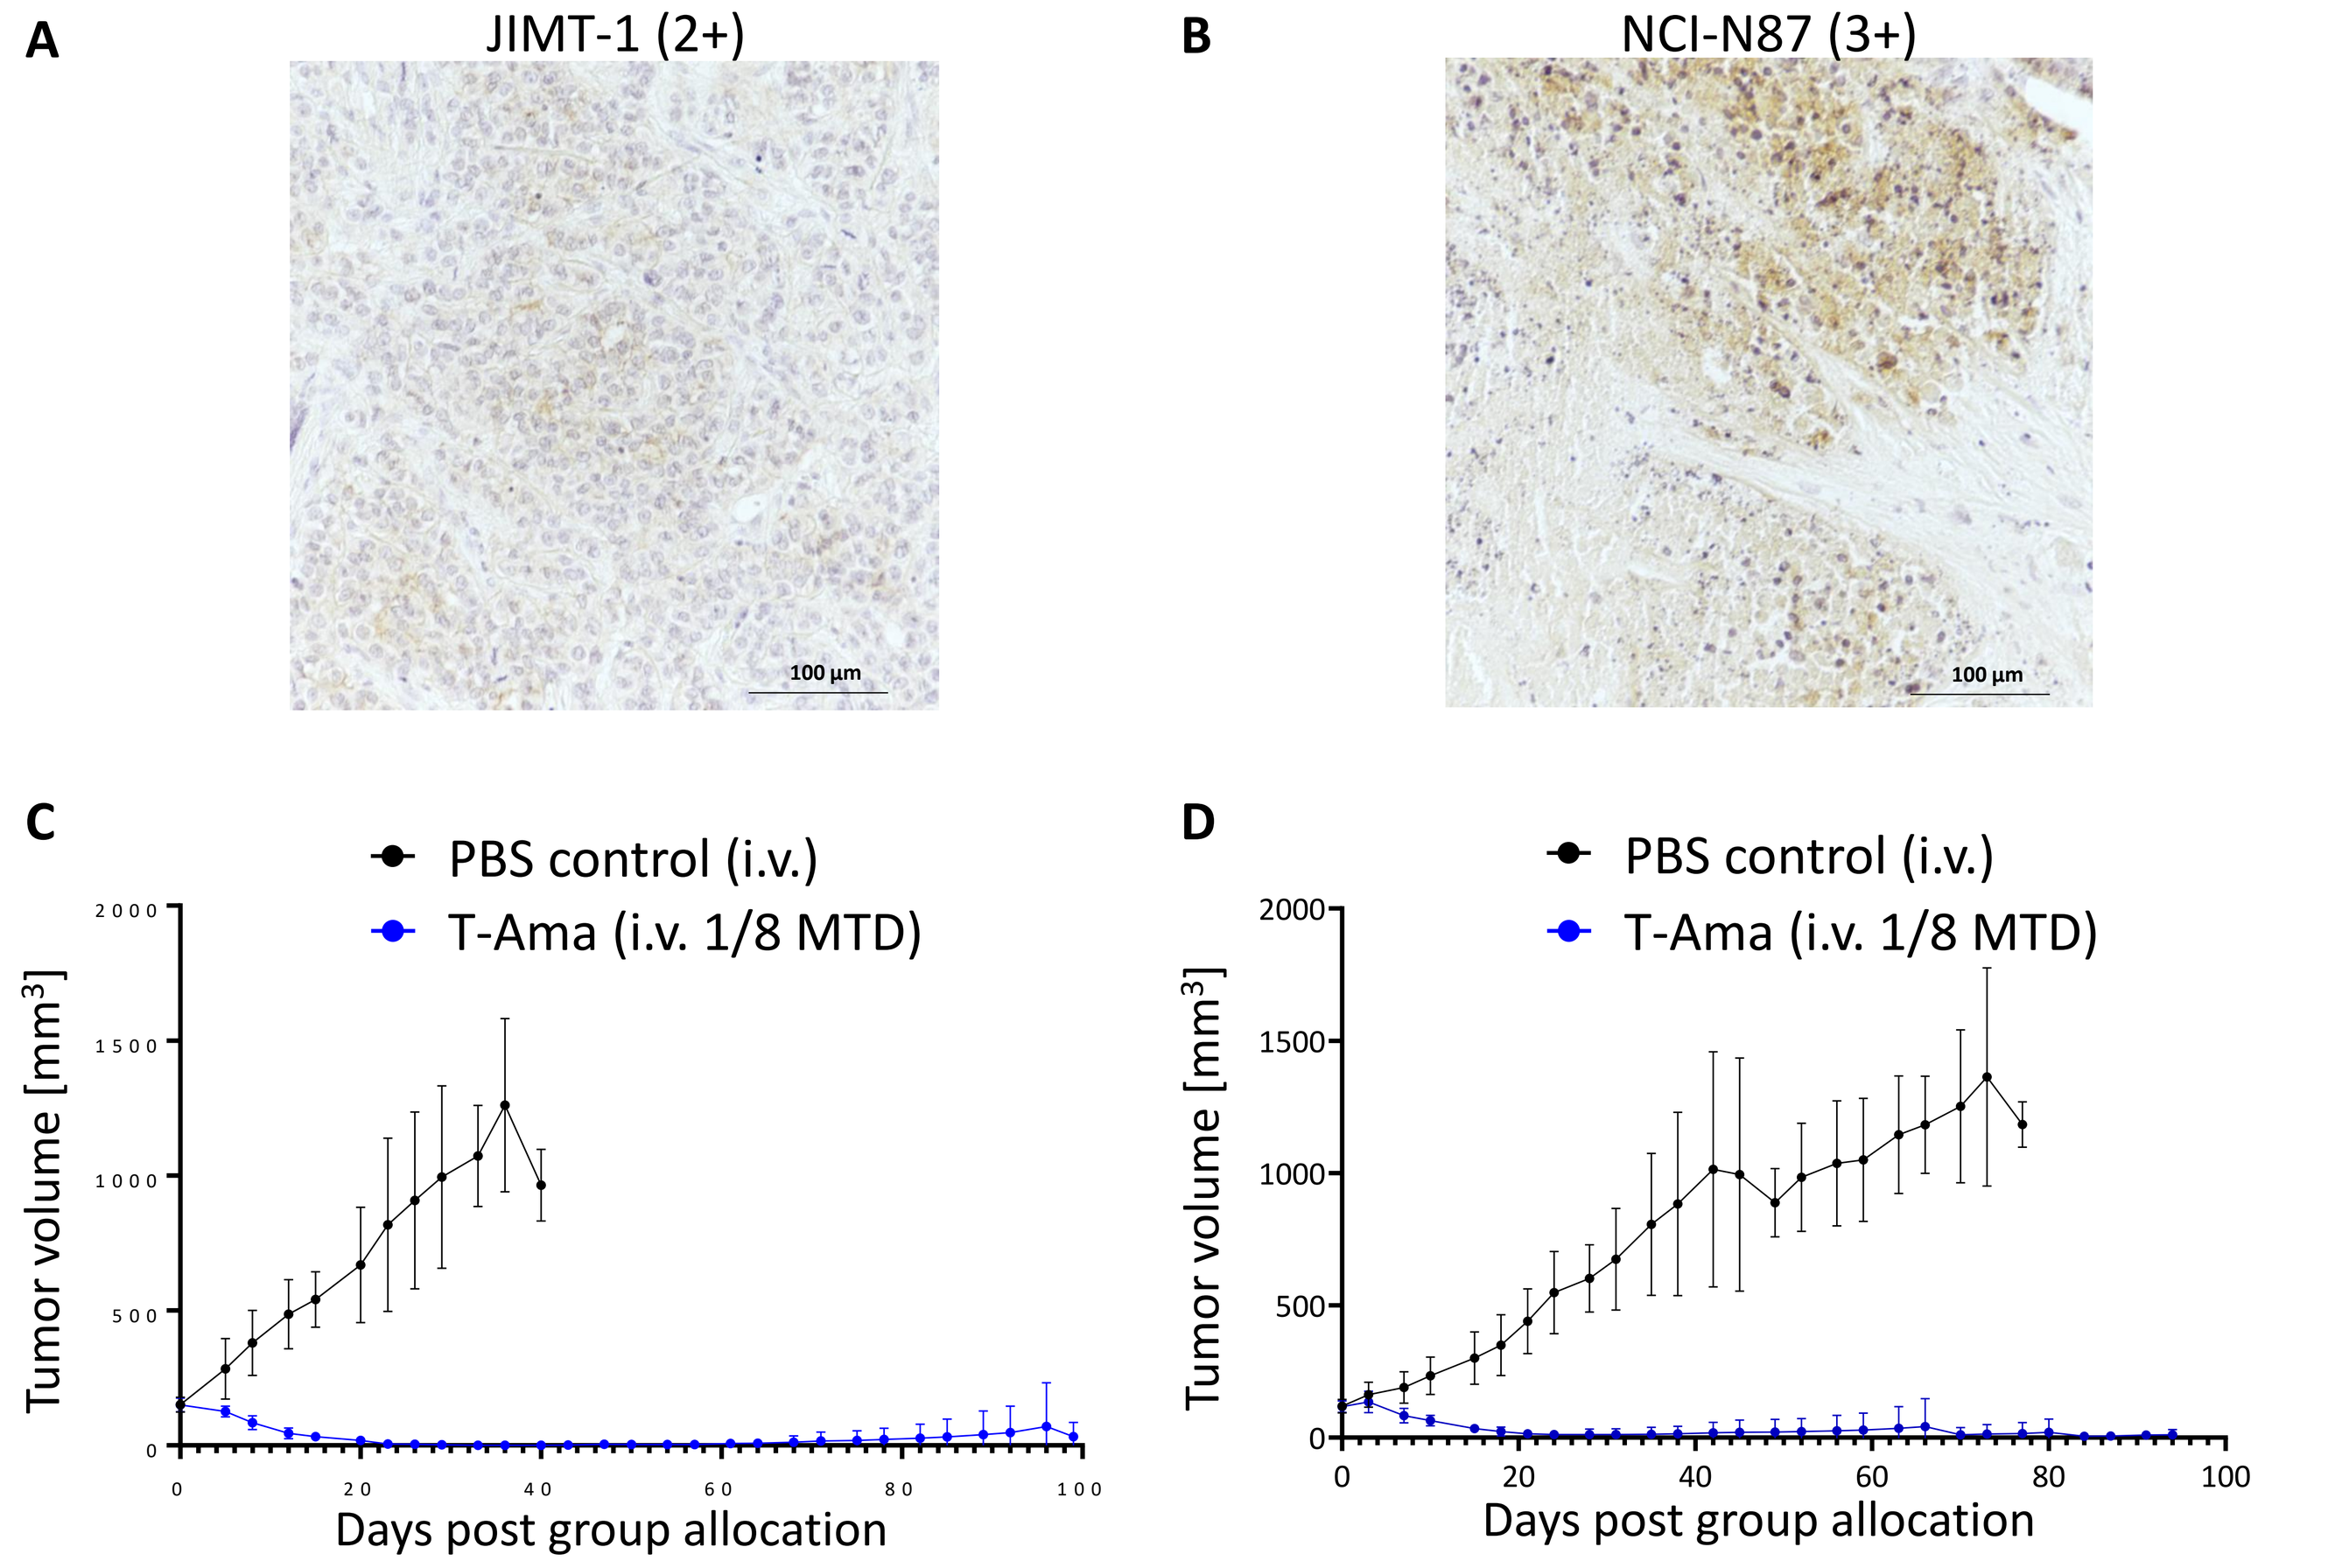
A
JIMT-1 (2+)
B
NCI-N87 (3+)
100 µm
C
D
PBS control (i.v.)
T-Ama (i.v. 1/8 MTD)
Tumor volume [mm³]
Days post group allocation

Figure 1: Anti-HER2 ATAC®-treatment led to CR in HER2⁺ breast and gastric cancer CDX models.
A-B: Immunohistochemistry staining for HER2 protein expression in a s.c. breast cancer JIMT-1 **(A)** and gastric cancer NCI-N87 **(B)** CDX tumor. **C-D:** Anti-tumor efficacy of an anti-HER2 ATAC® in breast cancer JIMT-1 **(C)** or gastric cancer NCI-N87 **(D)** CDX model. Mice were treated with either PBS (control) or an anti-HER2 ATAC® (n=10 mice, Mean +/- SD).

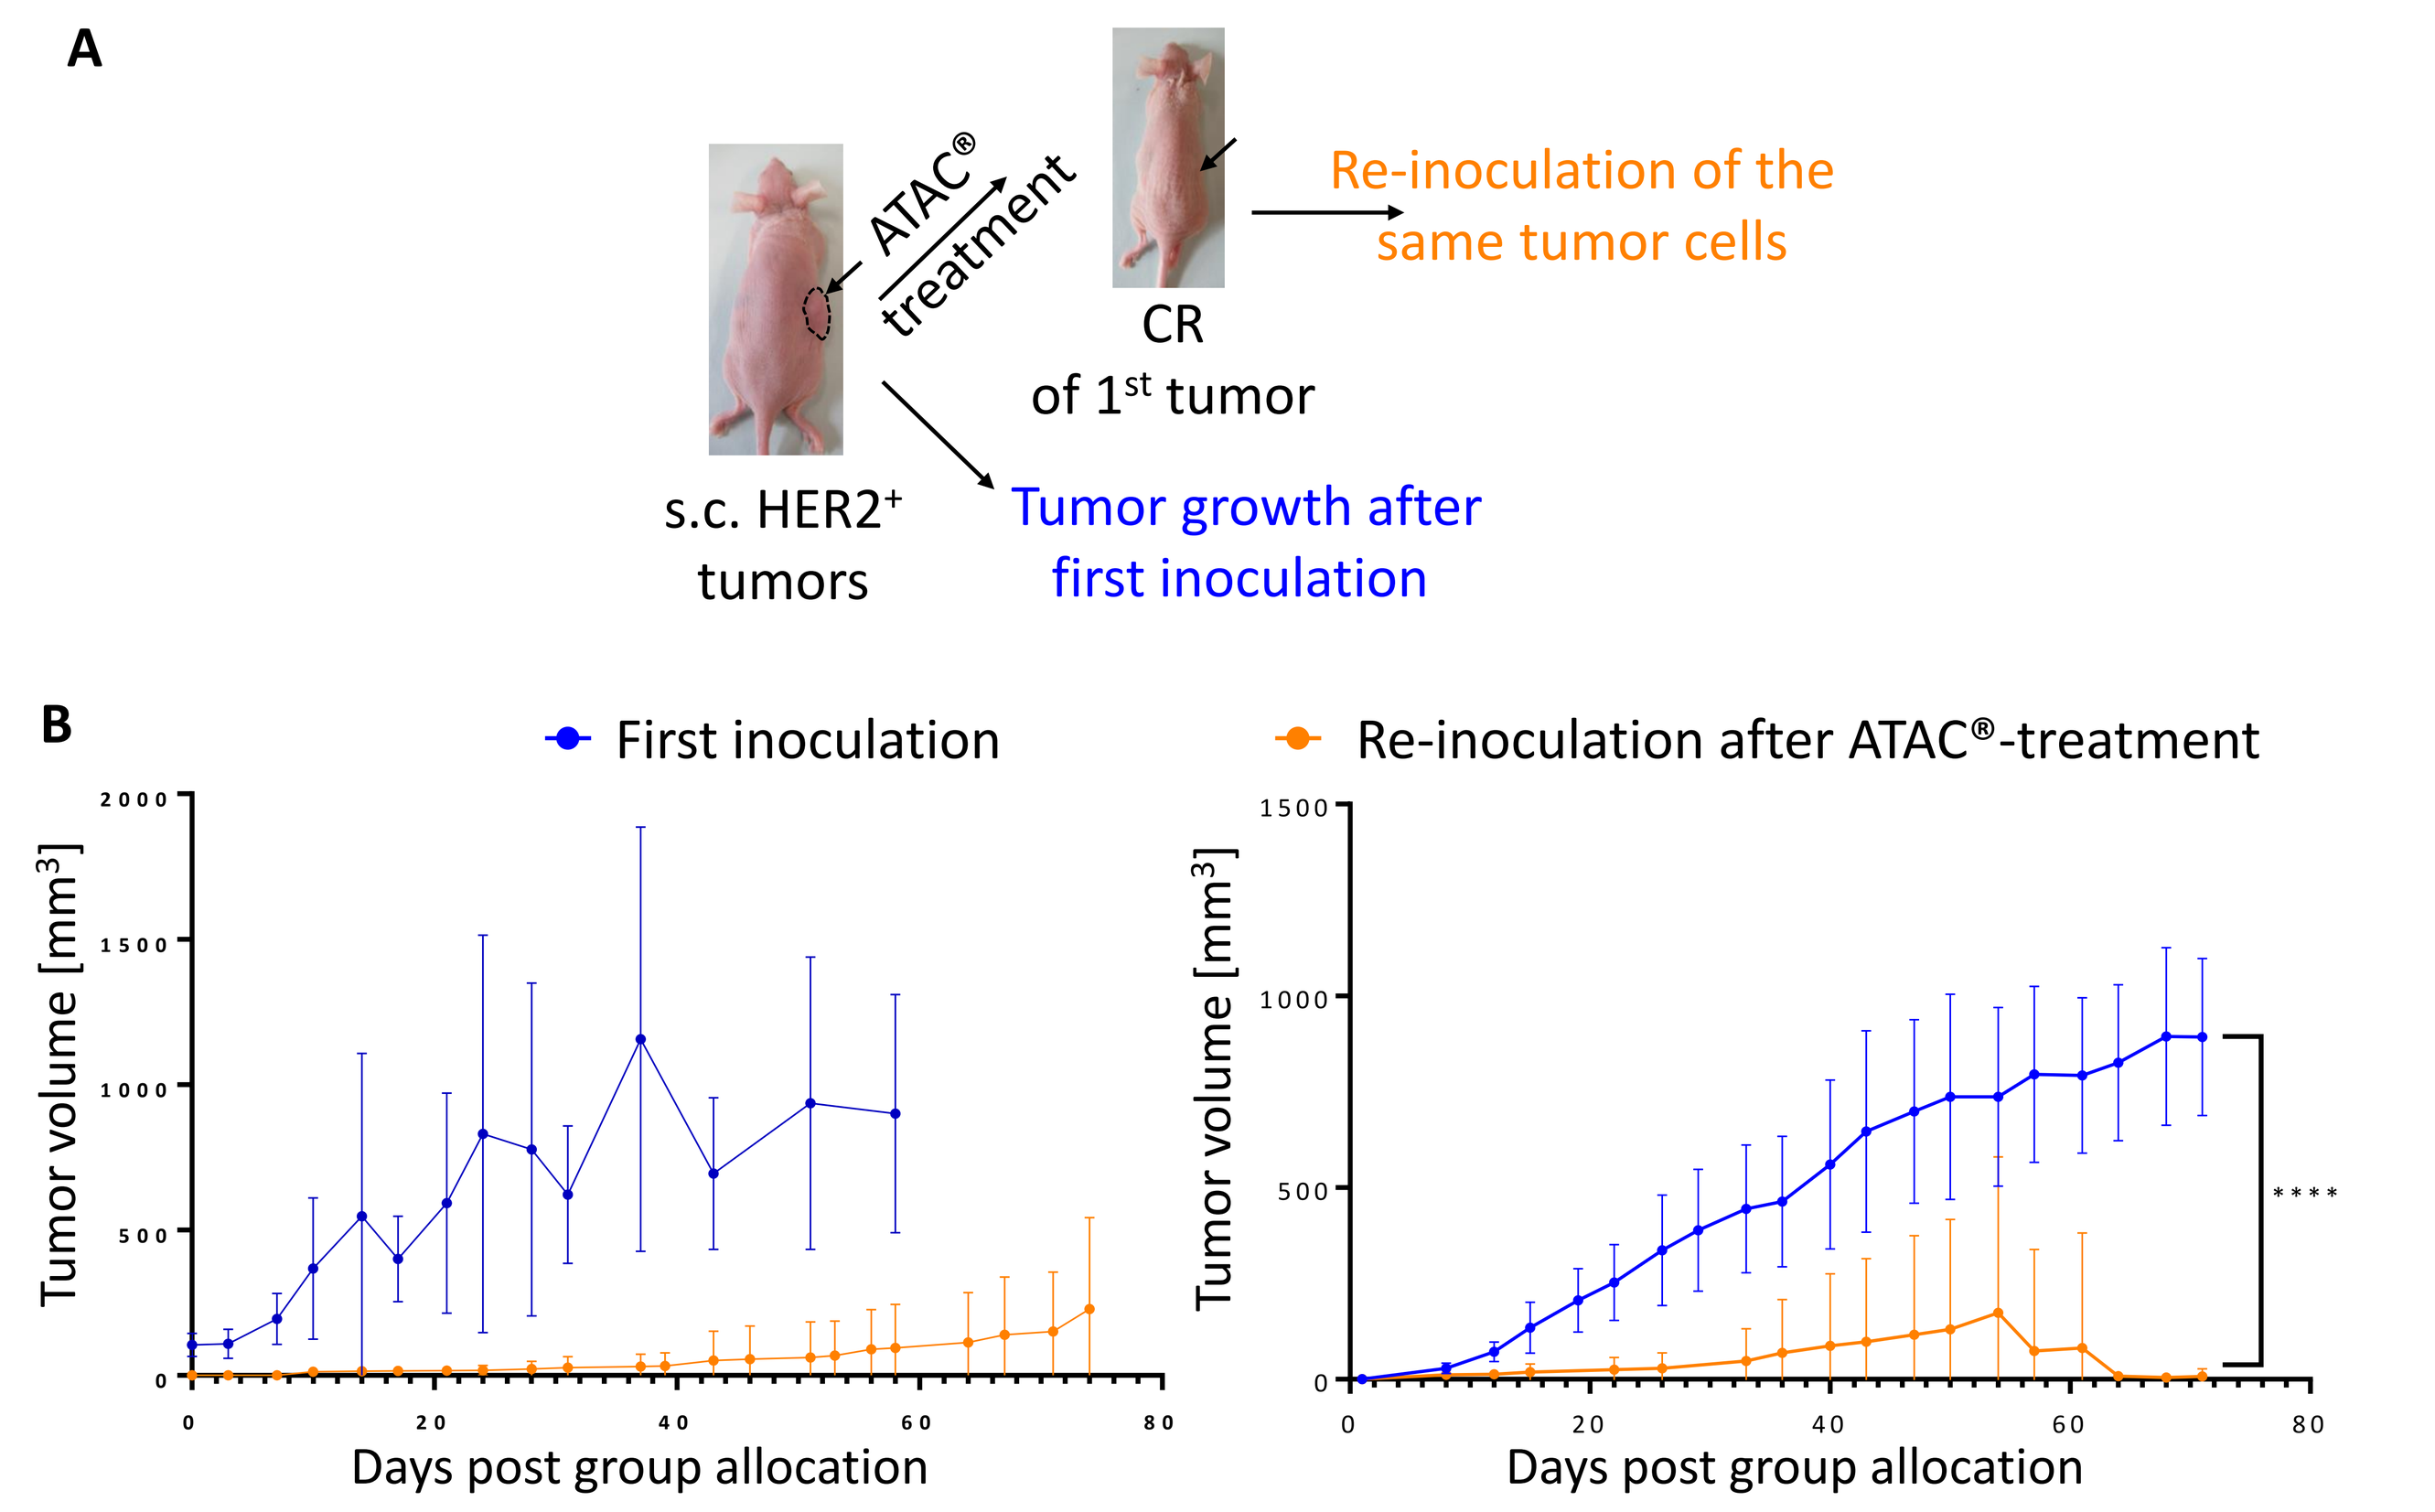
A
ATAC® treatment
CR
of 1st tumor
Re-inoculation of the same tumor cells
s.c. HER2⁺ tumors
Tumor growth after first inoculation
B
First inoculation
Re-inoculation after ATAC®-treatment
Tumor volume [mm³]
Days post group allocation

Figure 2: Anti-HER2 ATAC®-treatment resulted in significantly reduced tumor growth of HER2⁺ JIMT-1 and NCI-N87 cancer cells after re-inoculation.
A: Scheme of the experimental set-up of the re-challenge experiment. **B:** Tumor growth of JIMT-1 (left) or NCI-N87 (right) in untreated mice was compared to tumor growth in mice which were re-challenged with the same tumor after CR by ATAC®-treatment. (n=15 mice, Mean +/-SD; *: p-value <0.0001 (unpaired Welch t-test, Holm-Sidak correction)).

Treatment with anti-HER2 ATAC® induces immunogenic cell death in homogenous HER2⁺ cell line-derived and heterogenous HER2⁺ patient-derived xenograft models.

Based on the finding that ATAC®-treatment reduces take rate of the same tumor upon re-challenge, it was investigated whether ATAC®-based ADCs can activate the immune system. Consequently, ICD marker including calreticulin and HMGB1 were analyzed *in vitro* and *in vivo*. Treatment of NCI-N87 with anti-HER2 ATAC® led to increased apoptosis and surface expression of calreticulin (CRT) compared to untreated cells *in vitro* (Figure 3). This observation was confirmed in NCI-N87 CDX tumors *in vivo* (Figure 3). Interestingly, only in hIgG⁺ tumor regions HMGB1 expression (Figure 4) was observed in mice which were treated with anti-HER2-ATAC®. Additionally, single i.v. dose of an anti-HER2 ATAC® resulted in a significant tumor growth delay in a HER2 low expressing heterogeneous patient-derived xenograft (PDX) model of TNBC. Like CDX tumors, enhanced HMGB1 expression was observed in PDX tumors treated with anti-HER2 ATAC® (Figure 5).

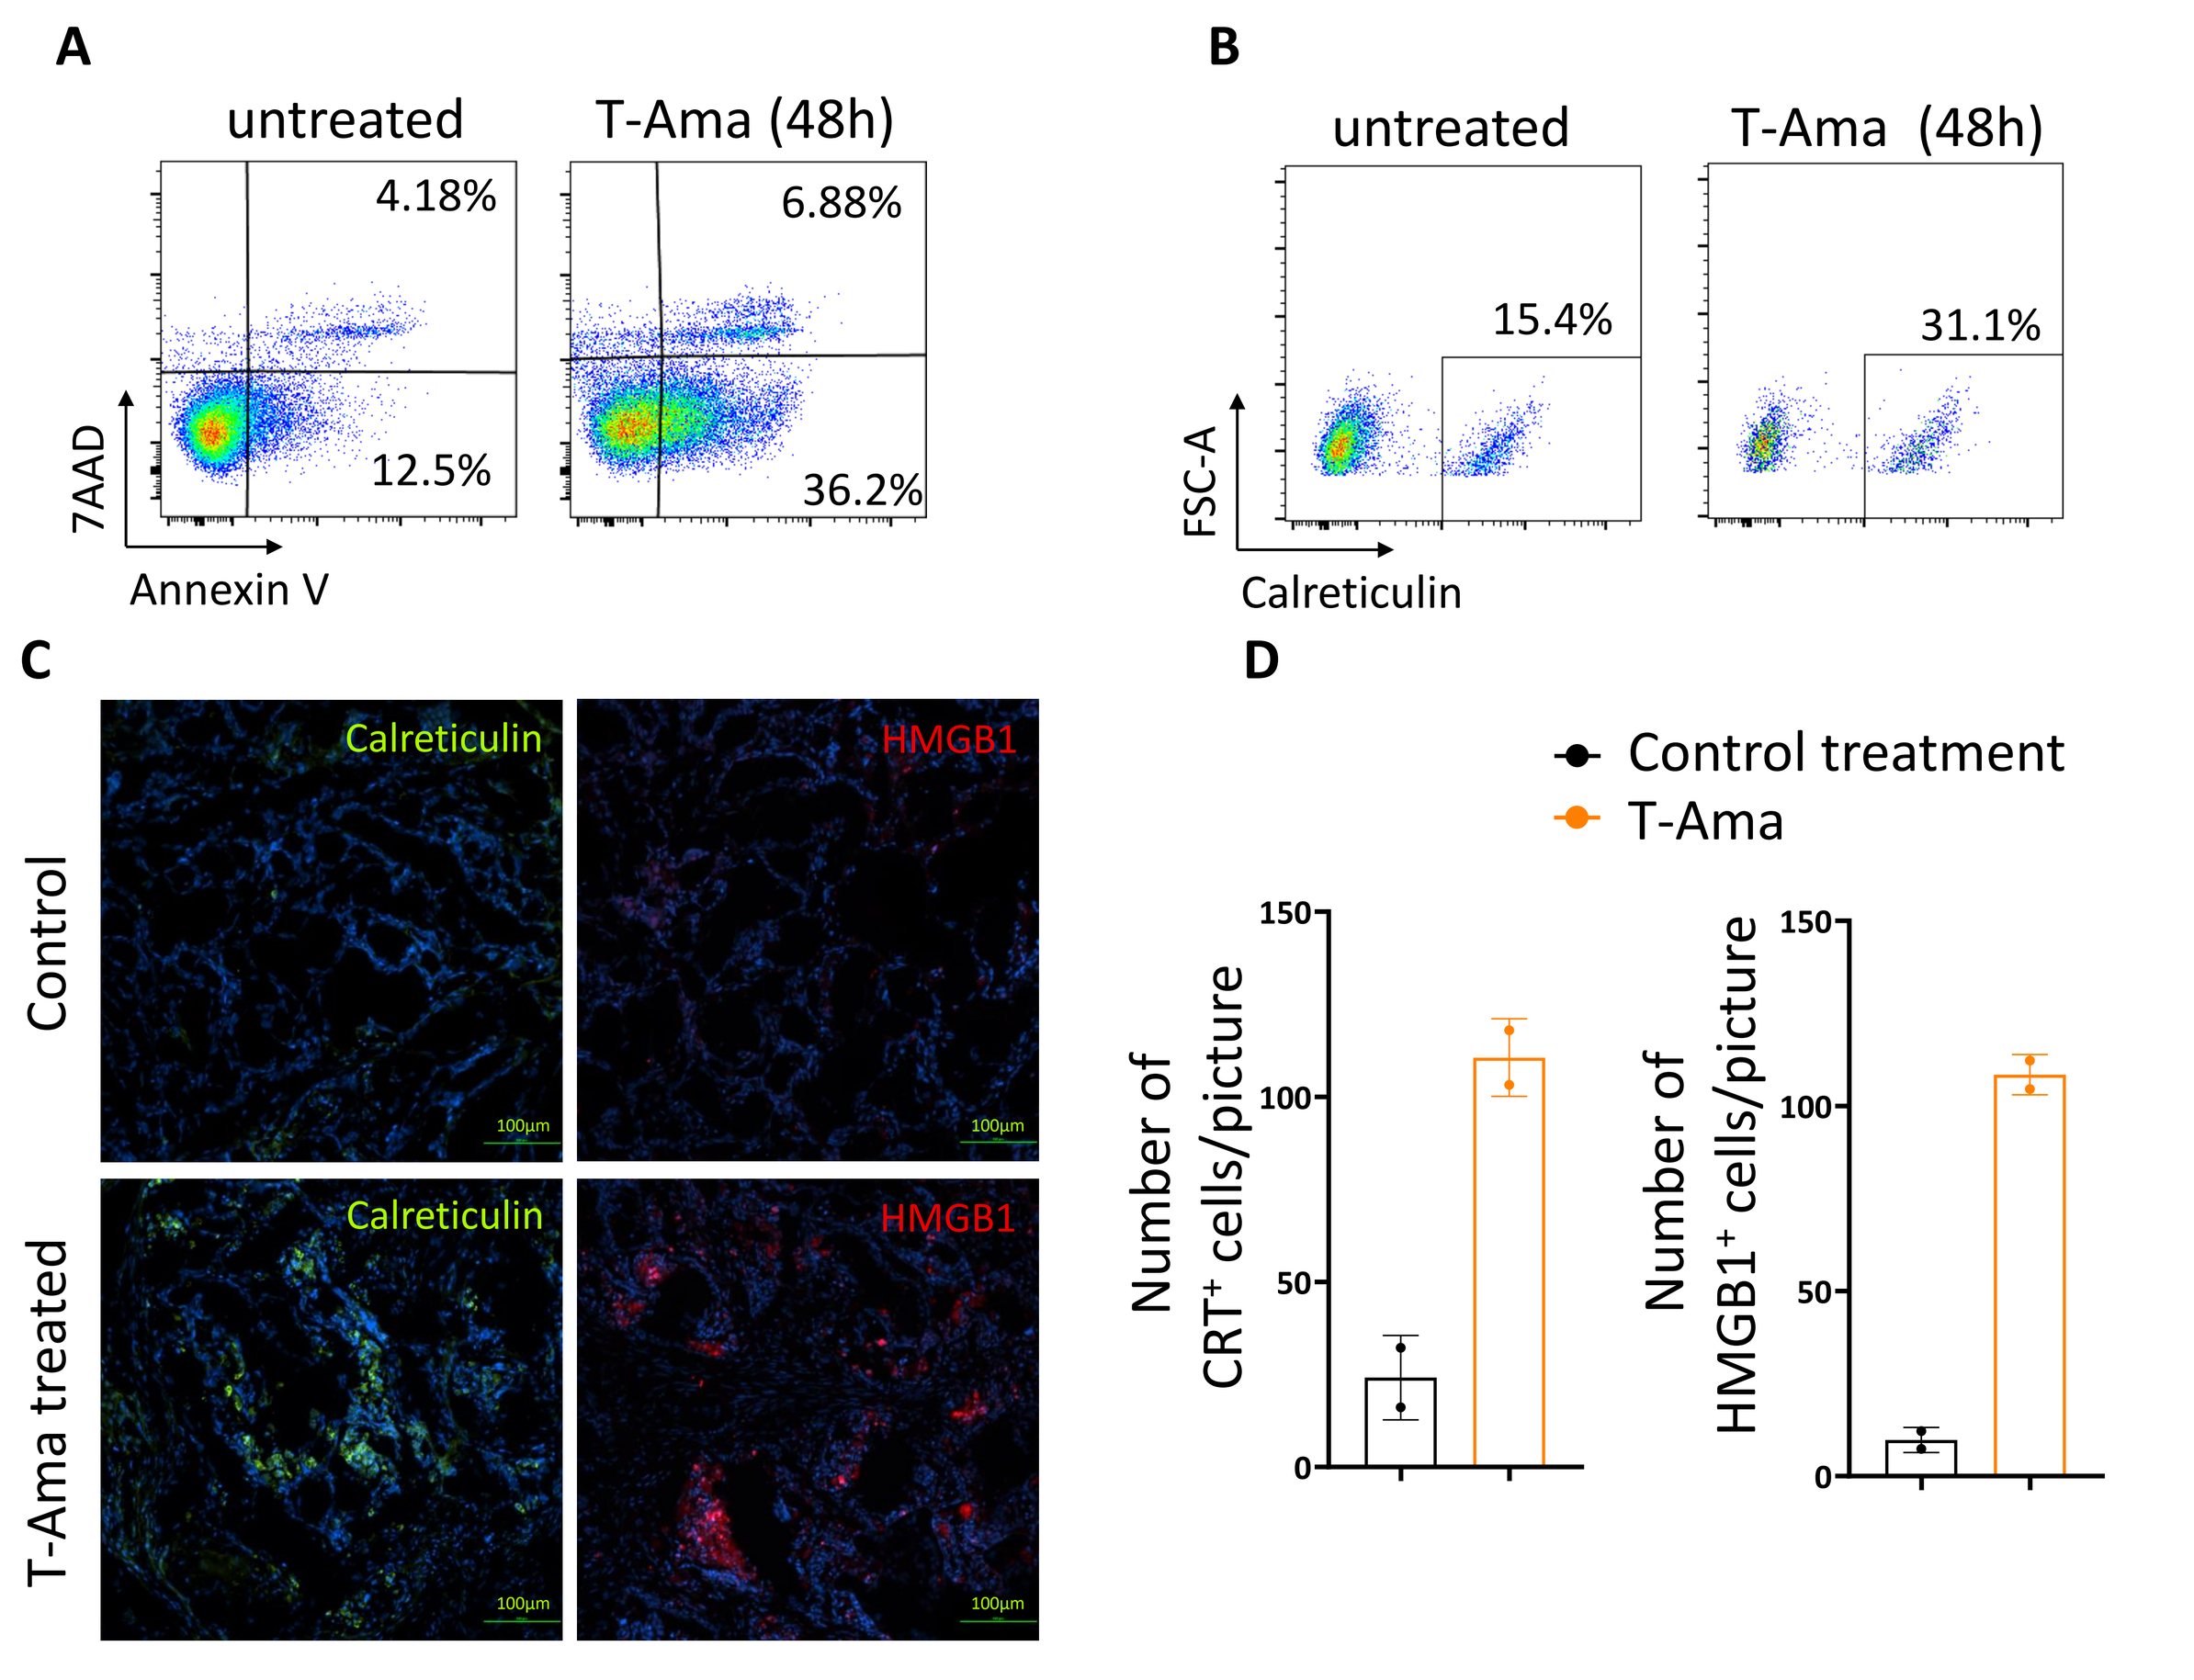
A
untreated
T-Ama (48h)
4.18%
12.5%
6.88%
36.2%
7AAD
Annexin V
B
untreated
T-Ama (48h)
15.4%
31.1%
FSC-A
Calreticulin
C
Control
T-Ama treated
Calreticulin
HMGB1
D
Control treatment
T-Ama
Number of CRT⁺ cells/picture
Number of HMGB1⁺ cells/picture

Figure 3: ATAC®-treatment induces apoptosis and triggers ICD *in vitro* and *in vivo*.
A: Early (Annexin V⁺/7AAD⁻) and late apoptosis (Annexin V⁺/7AAD⁺) of untreated or anti-HER2 ATAC®-treated NCI-N87 cells was determined by flow cytometry. **B:** Surface calreticulin expression on untreated or anti-HER2 ATAC®-treated NCI-N87 cells was analyzed by flow cytometry. **C-D:** IF staining **(C)** and statistical evaluation **(D)** of calreticulin (green) and HMGB1 (red) on NCI-N87 CDX tumors either anti-HER2 ATAC®-treated or untreated (n=2).

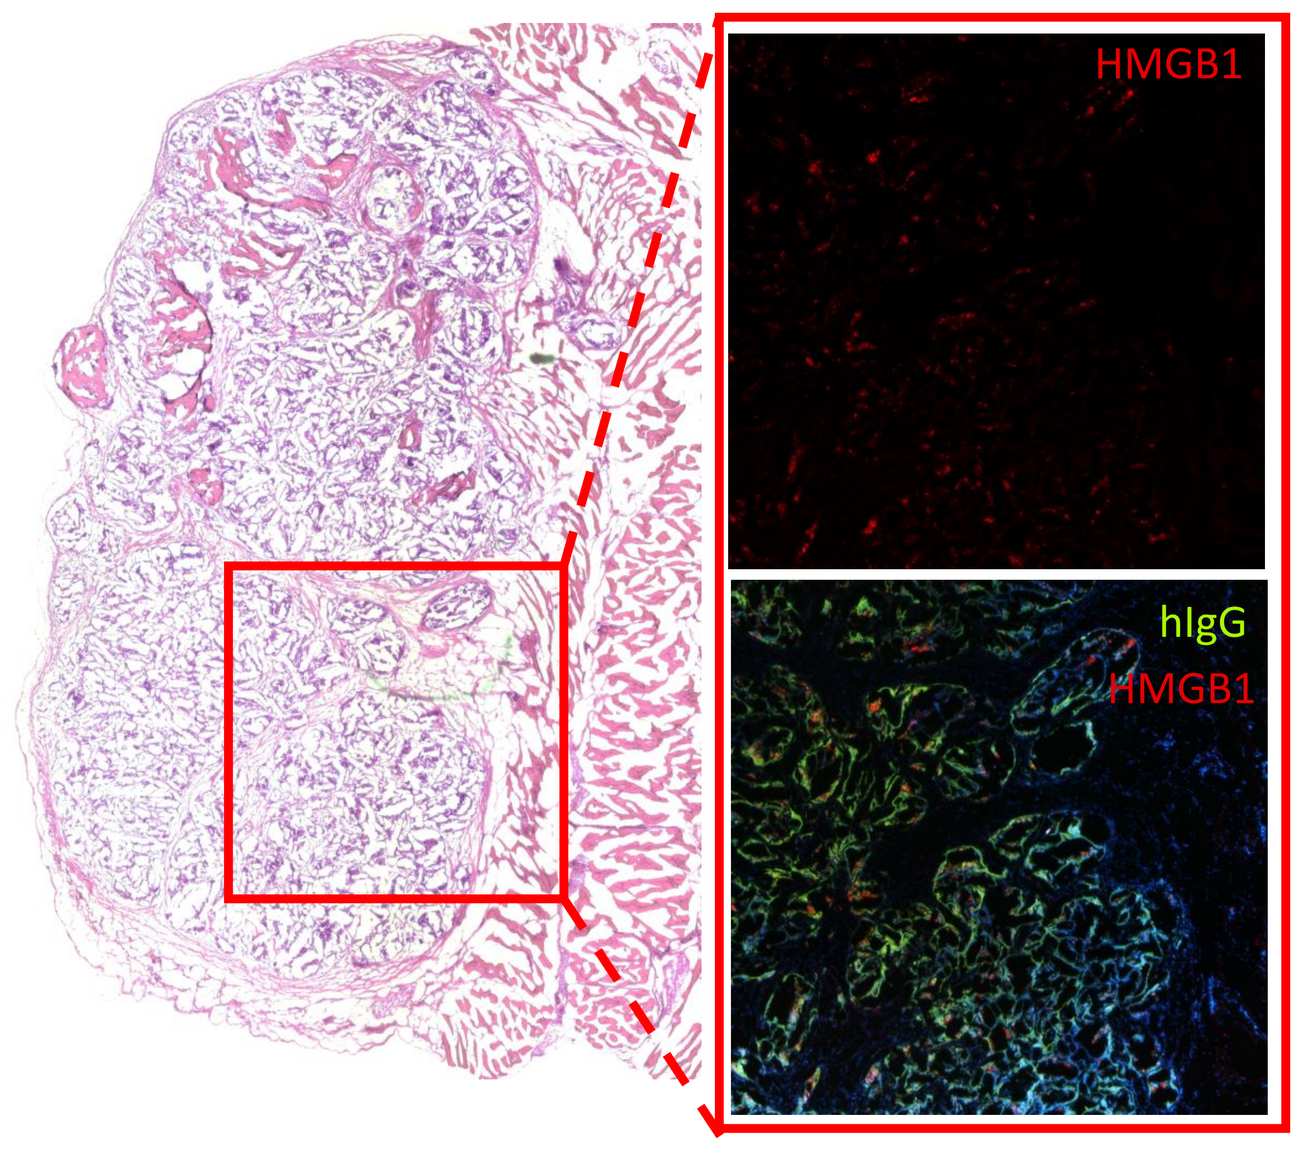
HMGB1
hIgG
HMGB1

Figure 4: ATAC®-treatment induces HMGB1 expression in hIgG⁺ tumor cells.
H&E staining of NCI-N87 CDX tumor treated with anti-HER2 ATAC®. Immunofluorescence staining of fresh frozen tumor sections for HMGB1 and human IgG (hIgG).

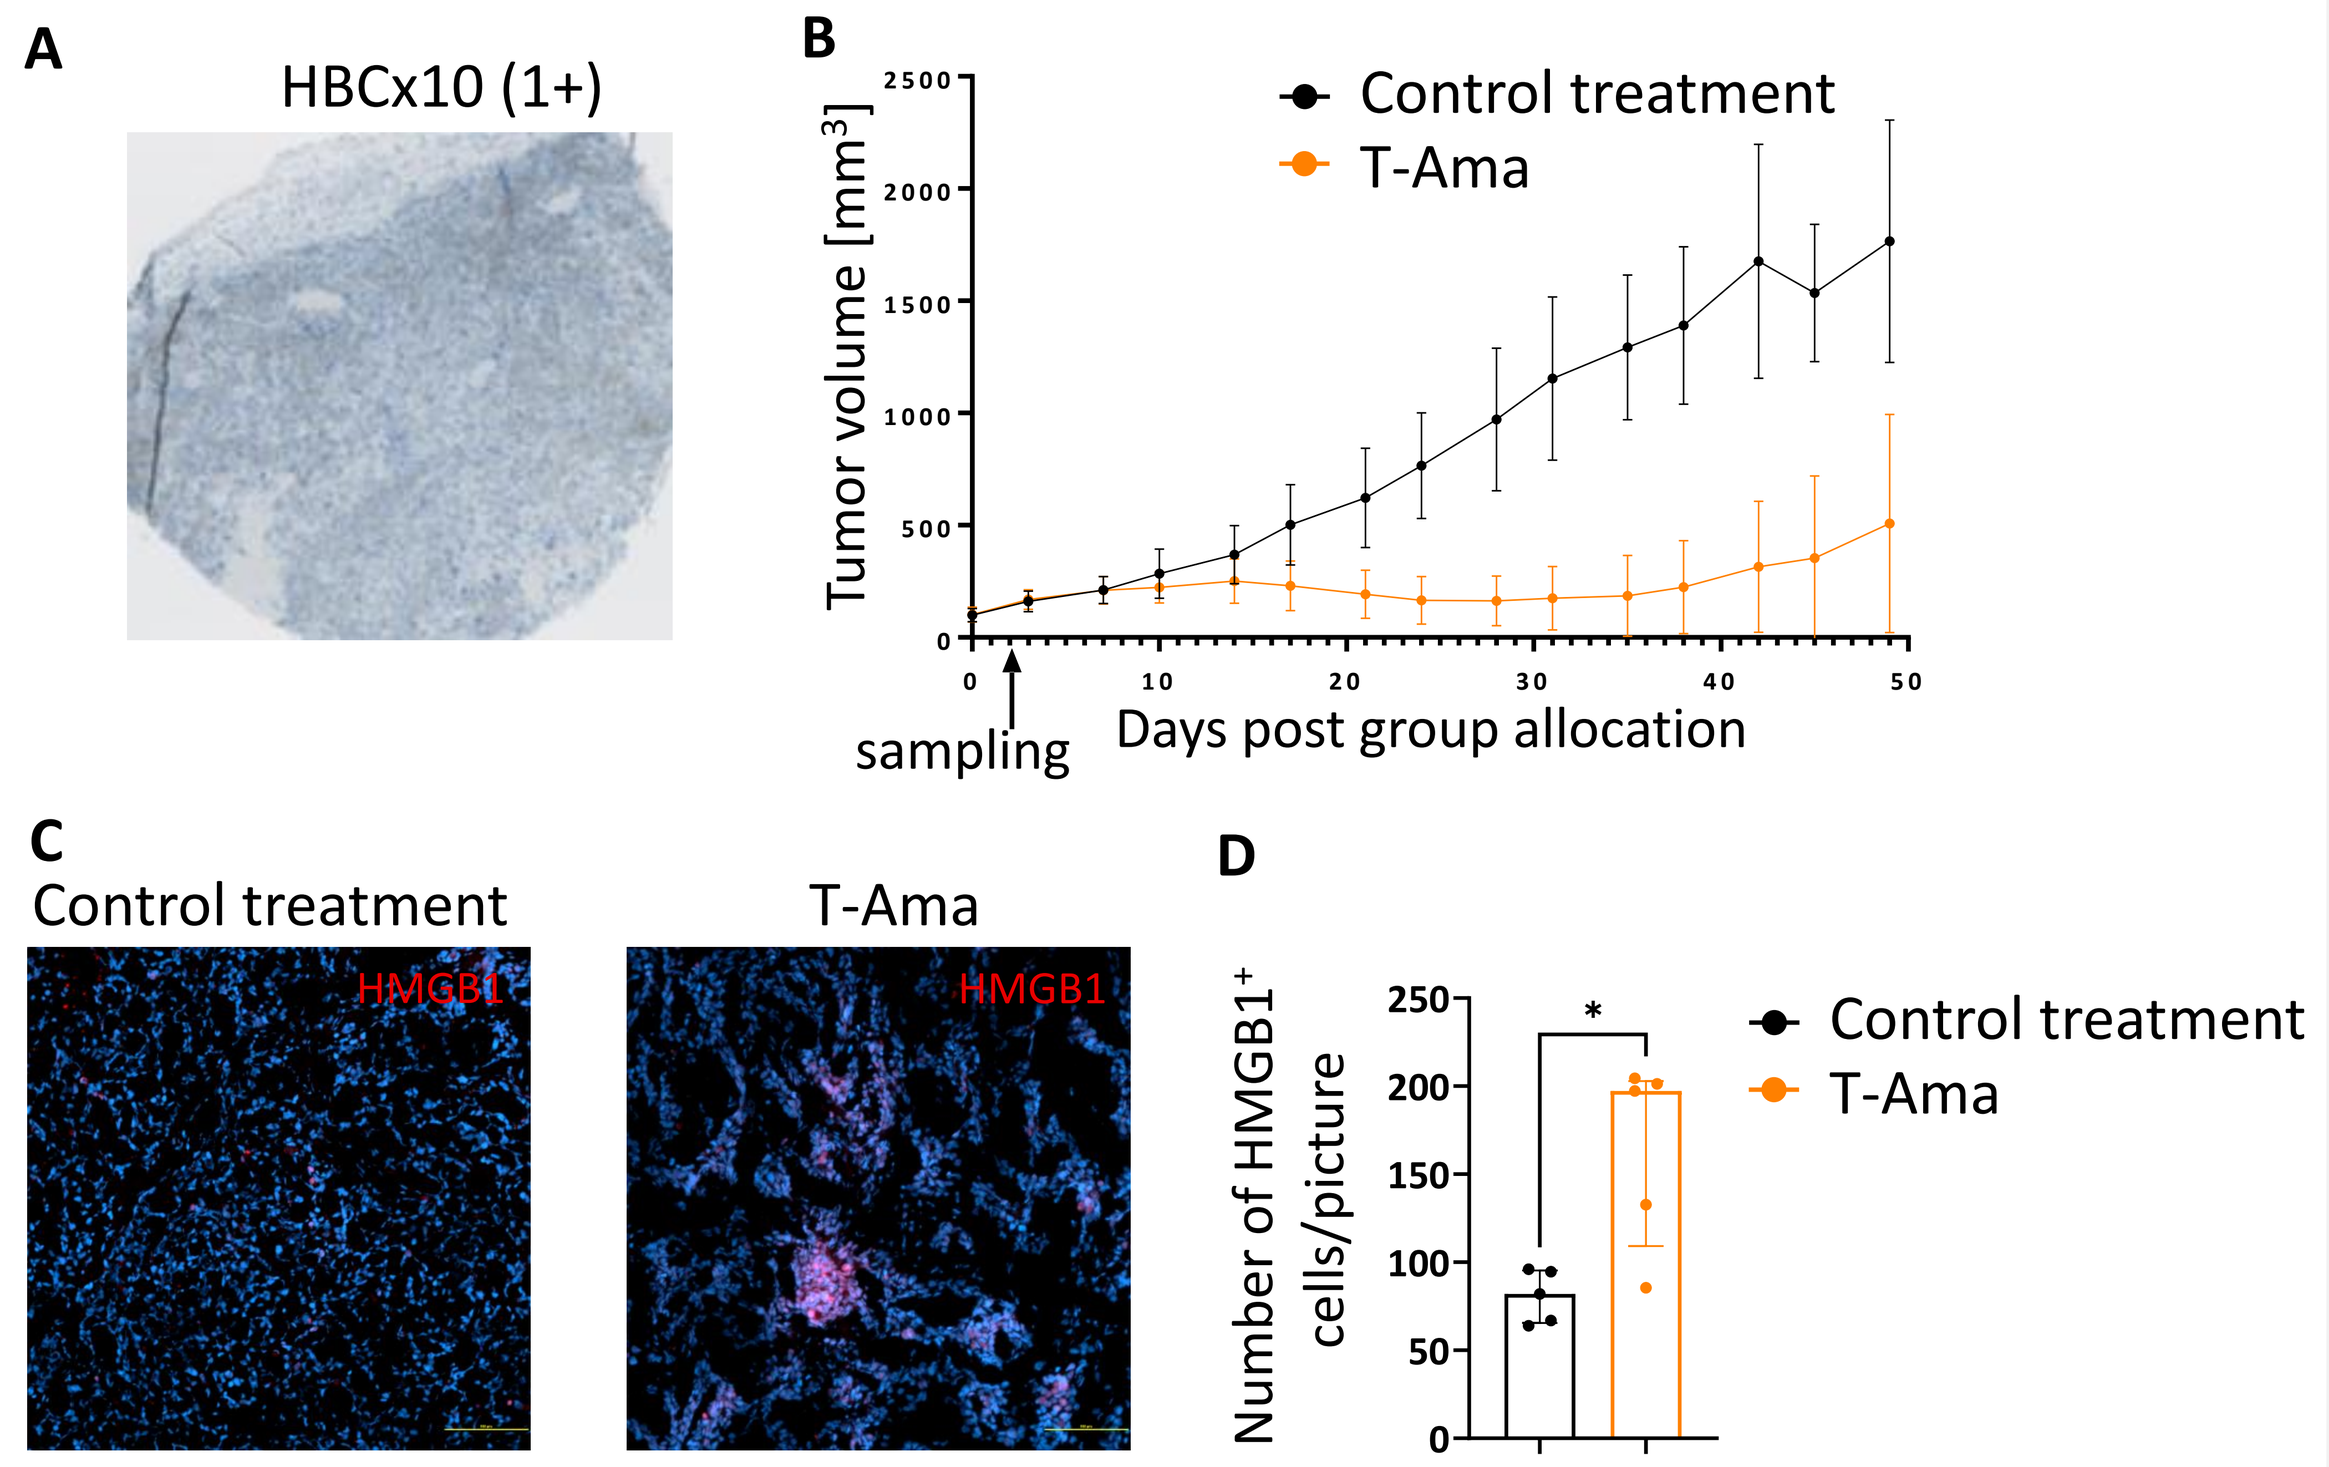
A
HBCx10 (1+)
B
Control treatment
T-Ama
Tumor volume [mm³]
sampling
Days post group allocation
C
Control treatment
T-Ama
HMGB1
D
Number of HMGB1⁺ cells/picture
*

Figure 5: Anti-HER2 ATAC®-treatment led to CR in a Her2 (1+) breast cancer PDX model.
A: IHC staining for HER2 protein expression in a s.c. TNBC PDX model. **B:** Anti-tumor efficacy of an anti-HER2 ATAC® in the same breast cancer PDX. Mice were treated with either non-targeting or an anti-HER2 ATAC® (n=10 mice, Mean +/- SD). IF staining **(C)** and statistical evaluation **(D)** of HMGB1 (red) expression on PDX tumors after the treatment with control ATAC or an anti-HER2 ATAC® (n=5).

3. Combined treatment of ATAC®-based ADCs with immune checkpoint inhibitors leads to a synergistic anti-tumor effect *in vivo*.

The immunity towards re-challenge of CDX tumors, the anti-tumor effect on heterogenous PDX model, together with the induction of ICD in CDX as well as PDX models suggest an activation of the immune system upon ATAC®-treatment. Consequently, it was investigated whether combination of ATAC®-based ADCs and ICIs (Ipilimumab, Pembrolizumab, Avelumab) may have a synergistic effect.

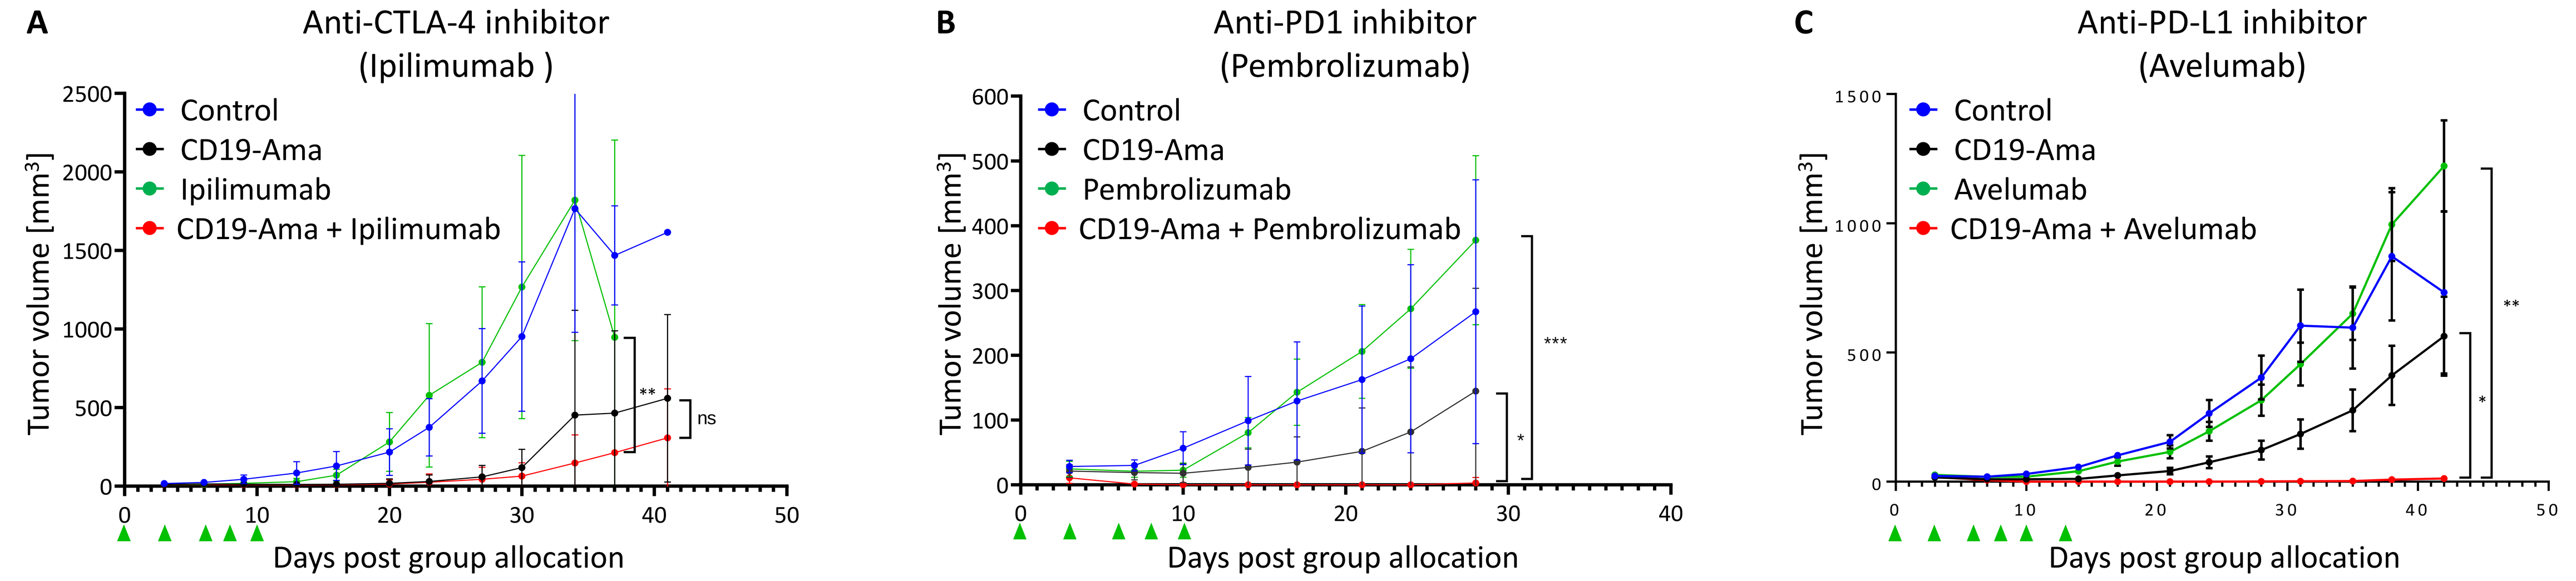
A
Anti-CTLA-4 inhibitor
(Ipilimumab)
Control
CD19-Ama
Ipilimumab
CD19-Ama + Ipilimumab
B
Anti-PD1 inhibitor
(Pembrolizumab)
Control
CD19-Ama
Pembrolizumab
CD19-Ama + Pembrolizumab
C
Anti-PD-L1 inhibitor
(Avelumab)
Control
CD19-Ama
Avelumab
CD19-Ama + Avelumab
Tumor volume [mm³]
Days post group allocation

Figure 6: Combined treatment with an ATAC®-based ADC and ICI results in a synergistic anti-tumor effect in a s.c. Burkitt lymphoma model.
A-C: Tumor growth of Raji cells inoculated s.c. in a 1:4 ratio with human PBMCs. Tumors were treated with PBS, an anti-CD19 ATAC®, Ipilimumab (q3d x 5) **(A)**, Pembrolizumab (q3d x 5) **(B)**, Avelumab (q3d x 6) **(C)** or in combination of ATAC®-based ADC and ICI . * p<0.05; ** p<0.001; *** p<0.0001 unpaired Welch t-test; Holm-Sidak correction.

Conclusion

The treatment with ATAC®-based ADCs led to complete and stable tumor remission in different cell line-derived xenograft models with homogenous antigen expression. The induced immunity in mice that achieved complete and stable tumor remission upon ATAC®-treatment suggest an activation of the immune system.

The complete remission of heterogenous patient-derived xenograft tumors which were treated with ATAC®-based ADCs further underline the involvement of the immune response because previous in-house studies already showed that ATAC®-based ADCs do not have any bystander effect upon extracellular drug-release.

This observation was accompanied by the finding of enhanced ICD marker expression *in vitro* and *in vivo*. Especially, the colocalization of hIgG⁺ tumor cells and HMGB1 staining highlights the correlation between the treatment with ATAC®-based ADCs and the induction of ICD.

Finally, the synergistic effect *in vivo* of ATAC®-based ADCs and ICIs support the previous stated link between the activation of the immune system due to ATAC®-treatment.

Together, the presented data highlights the general concept of the synergistic effect of ATAC-based ADCs® and ICIs which applies to several types of ICIs thus strengthening the scientific rationale for combination treatments in clinical trials.

References

(1) Anderl J, Echner H, Faulstich H. Chemical modification allows phallotoxins and amatoxins to be used as tools in cell biology. Beilstein J Org Chem. 2012; 8:2072-84

(2) Moldenhauer G, Salnitov AV, Lüttgau S et al. Therapeutic potential of amanitin-conjugated anti-epithelial cell adhesion molecule monoclonal antibody against pancreatic carcinoma. J Natl Cancer Inst. 2012; 104(8):622-34

(3) Liu Y, Zhang X, Han C, et al. TP53 loss creates therapeutic vulnerability in colorectal cancer. Nature 2015; 520(7549):697-701

ATAC® is a registered EU trademark of Heidelberg Pharma Research GmbH, No. 017988594

Acknowledgements

We thank Daniela Petzinger and Claudia Dinkelacker for the establishment of IHC and IF stainings on human tissue sections and *in vitro* cell culture assays.

Heidelberg Pharma Research GmbH
Gregor-Mendel Straße 22
68526 Ladenburg
Germany

Phone: +49-6203-1009 0

Email: info@hdpharma.com
https://www.heidelberg-pharma.com