Subcutaneous dosing increases the therapeutic index of Amatoxin-based ADCs

#1523

AACR Annual Meeting 2023

Kristin Decker, Marija Vranic, Marisa Schmitt, Irina Dranova, Anikó Pálfi, Andreas Pahl, Torsten Hechler and Michael Kulke
Heidelberg Pharma Research GmbH, Gregor-Mendel-Straße 22, 68526 Ladenburg, Germany

Heidelberg PHARMA
Focused Cancer Therapies

Introduction

Amatoxin-based antibody-drug conjugates (so called ATACs) are a new class of ADCs that are based on the ATAC® technology. They distinguish themselves from other ADCs by the use of the RNA polymerase II inhibitor amanitin as toxic payload[1, 2]. Currently HDP-101[3], an anti-BCMA ATAC is being tested in a first-in-human phase I/II clinical trial in multiple myeloma patients. In this trial HDP-101 is given as intravenous (i.v.) infusion as it is the standard administration route for ADCs.
The choice of the administration route depends on several factors including the convenience for the patient and the pharmacokinetic and –dynamic properties of the drug[4]. Each administration route has its own advantages and disadvantages. i.v. administrations leads to a rapid onset of action and almost 100% bioavailability, but it might also be painful, requires hospitalization and leads to high peak serum levels (Cmax) that can trigger toxicity. Subcutaneous (s.c.) administration, which is often used for antibody agents, on the other hand, has the advantage that the administration is very easy and can even be done by the patients themselves. Disadvantages are that the absorption rate is difficult to control and it may lead to local irritations[4]. In the end, the ideal administration route has to be determined empirically for every drug. Thus, in this study i.v. and s.c. administration of ATAC®-based ADCs was directly compared regarding tolerability, pharmacokinetics (PK) and anti-tumor efficacy. We show that s.c. dosing is a promising alternative to i.v. dosing for ATACs as it increases the therapeutic index (TI).

Methods

ATACs: anti-DIG, anti-CD37 and anti-PSMA antibodies, produced by Heidelberg Pharma (HDP) were conjugated site-specifically (THIOMAB® strategy) to cysteine-reactive amanitin-linker constructs, synthesized at HDP. Drug-antibody ratio according to LC-MS analysis was ~2.0 amanitins per IgG.

Mouse tolerability study: male CB17-SCID mice were treated with anti-DIG or anti-PSMA ATAC or PBS as single i.v. or s.c. dose on day 0. Doses were adjusted until the maximal tolerated dose (MTD) was reached.

Mouse PK studies: male CB17-Scid mice were treated with a single dose of ATAC s.c. or i.v.. Blood samples were taken at different time points, serum was generated, and ADC levels were measured by ELISA.

Efficacy studies: prostate model: 2.5x10^6 C4.2 cells were implanted s.c. into male CB17-SCID mice. At a mean tumor volume (TV) of 150 mm^3, mice were randomized (n = 10) and treated with a single i.v. or s.c. dose of anti-PSMA-Ama1 (HDP-103) or PBS. B-cell leukemia model: 2.5x10^6 MEC-2 cells were injected i.v. into female CB17-SCID mice. On day 3 the mice were treated with a single i.v. or s.c. dose anti-CD37-Ama1 or PBS.

Studies in Cynomolgus monkeys: 2-to-4-year-old monkeys were treated with a single i.v. or s.c. dose of Anti-PSMA-Ama1 in an escalating dose regiment. AST and ALT levels in the serum were measured over time using the methods recommended by the IFCC (at Wuxi).

Results

1. S.c. administration changes the pharmacokinetic profile and improves the tolerability of ATAC®-based ADCs in mice

The PK profile directly influences the tolerability and efficacy of a drug as it determines the maximal serum levels (Cmax), its serum half-life and the dose available over time (AUC). Therefore, the effect of s.c. dosing on the PK profile of ATAC®-based ADCs was investigated after a single i.v. or s.c. dose in mice (Figure 1).

A)

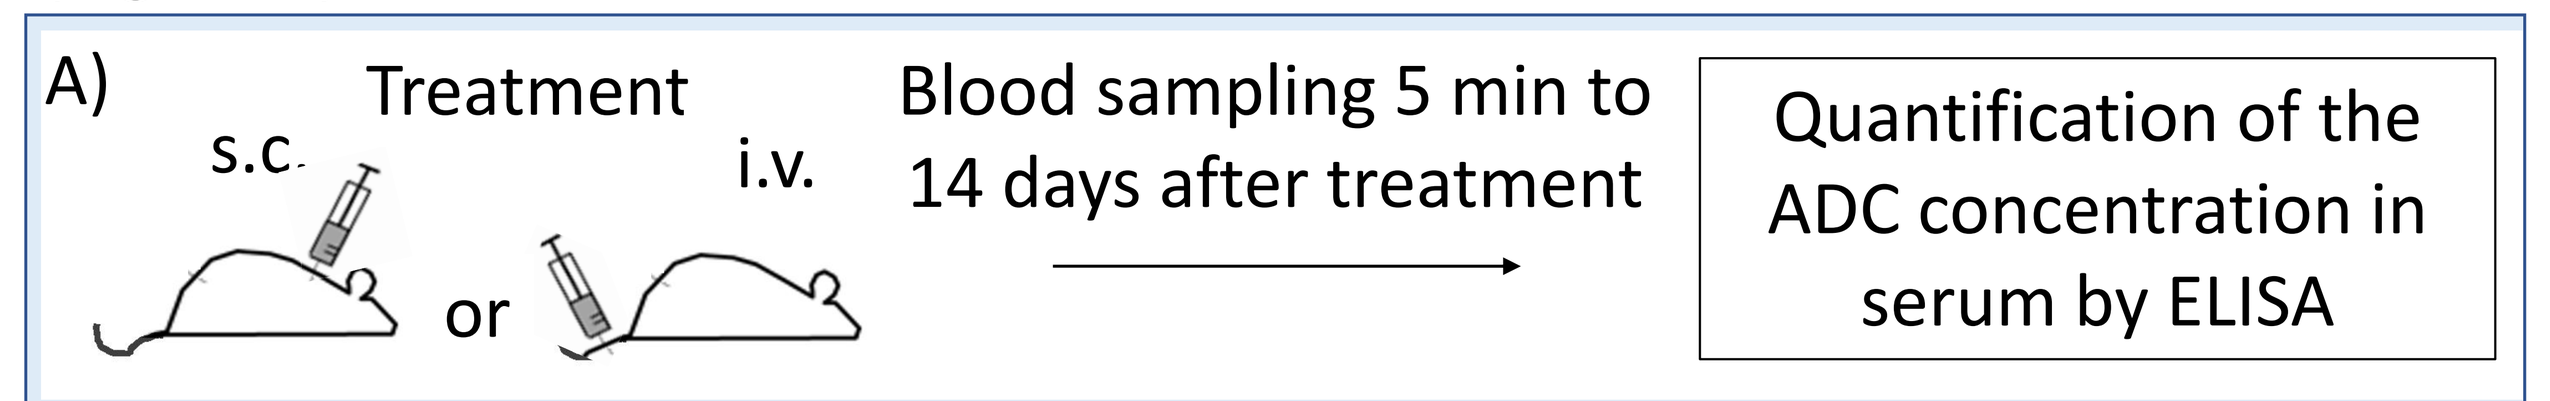
Treatment
s.c.
i.v.
or
Blood sampling 5 min to 14 days after treatment
Quantification of the ADC concentration in serum by ELISA

B)

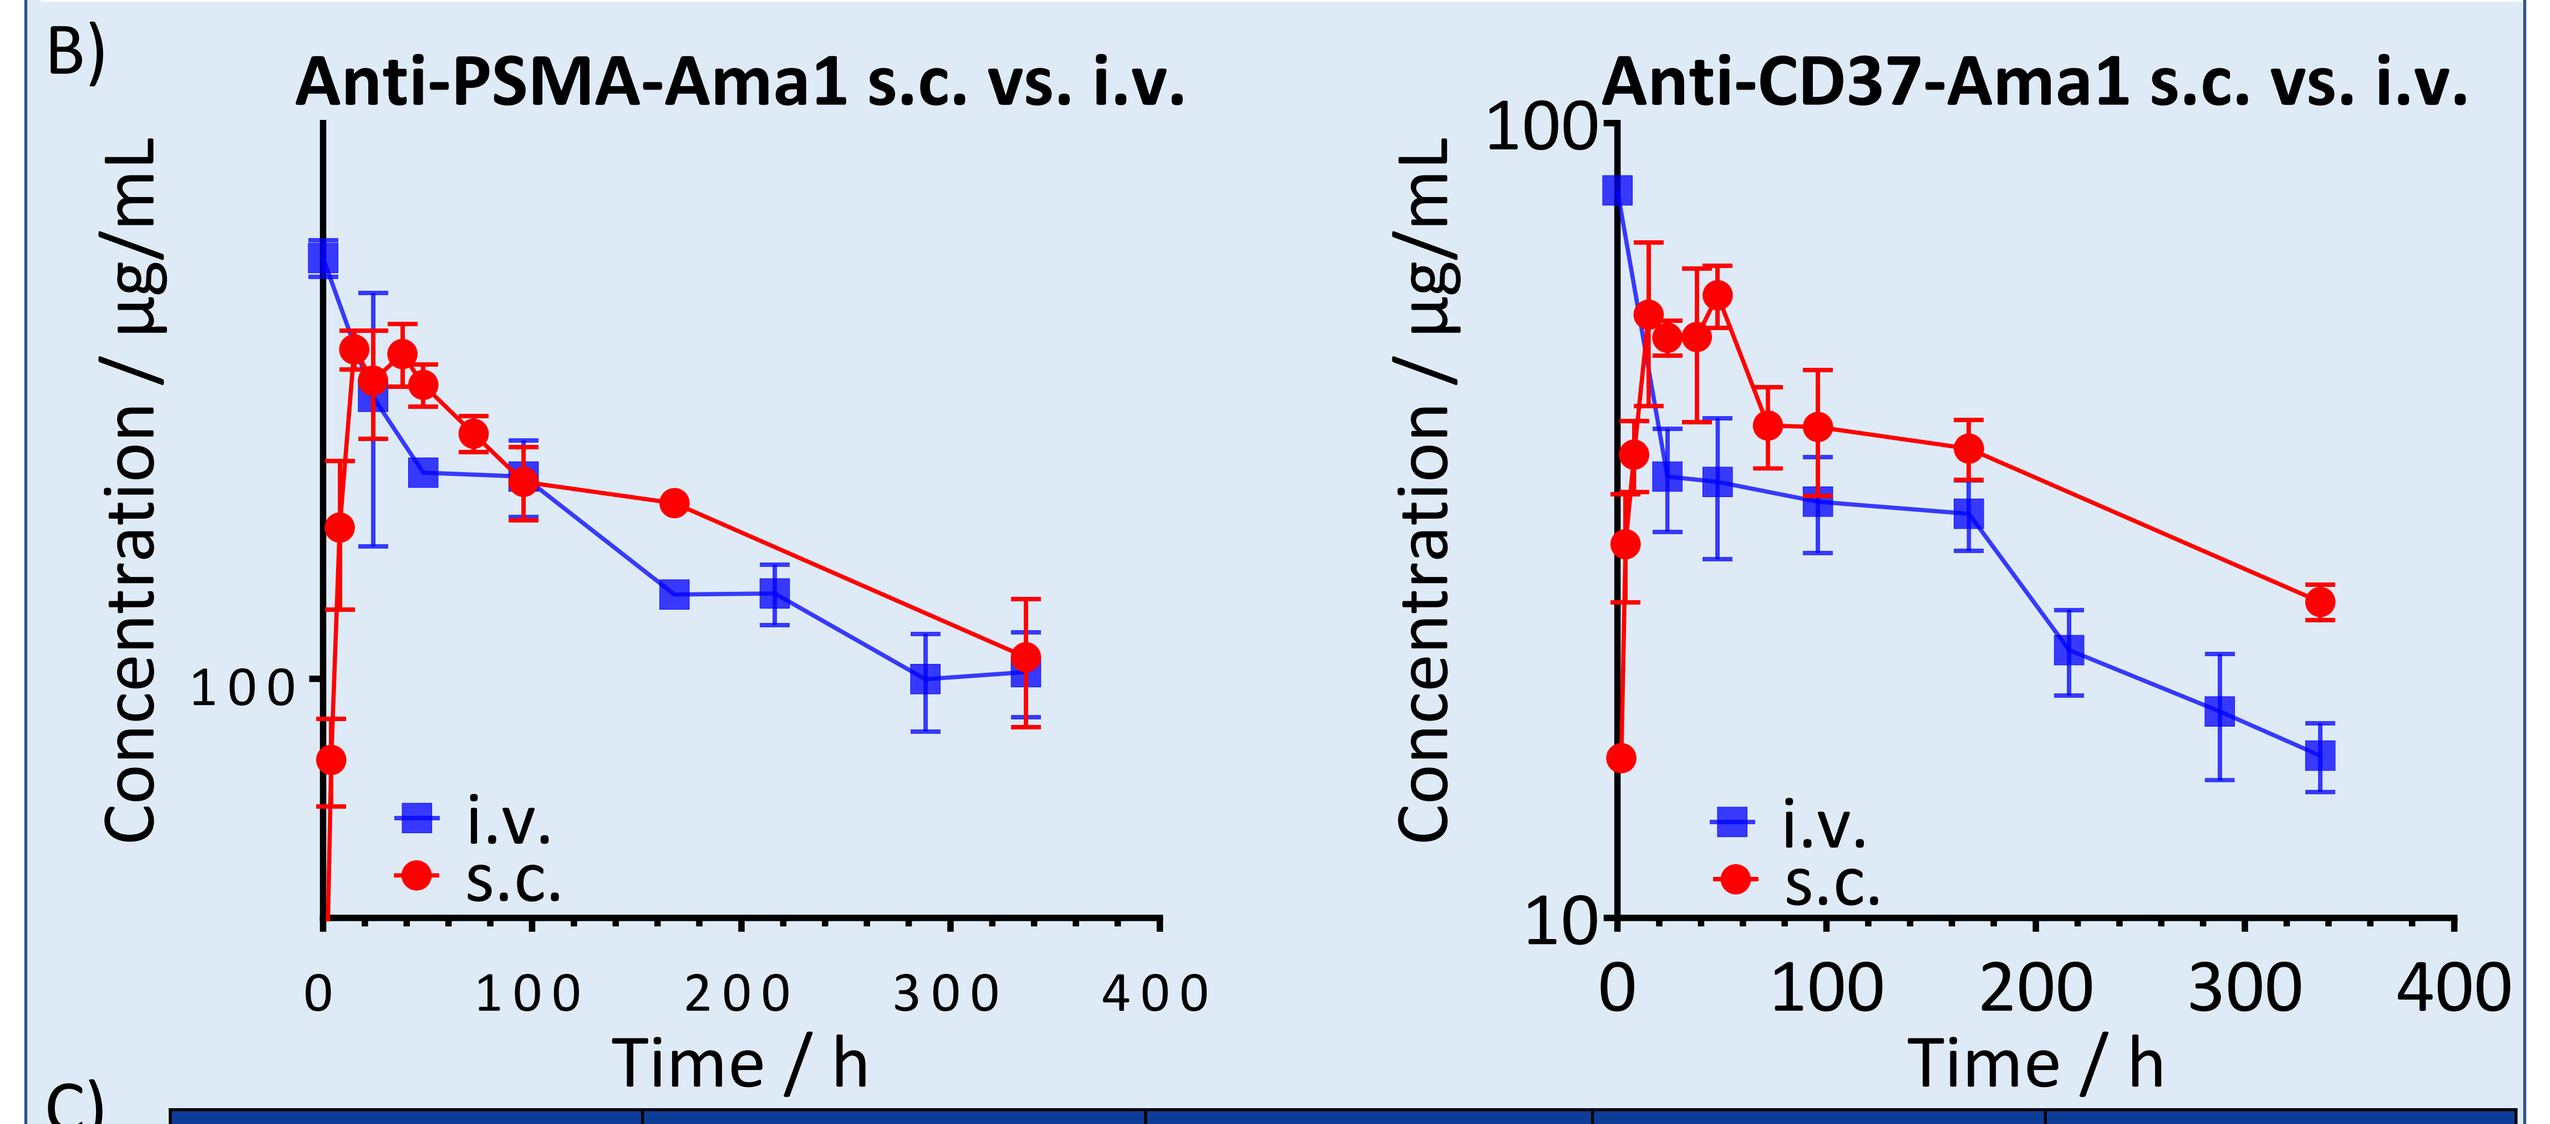
Anti-PSMA-Ama1 s.c. vs. i.v.
Anti-CD37-Ama1 s.c. vs. i.v.
Concentration / µg/mL
Time / h
i.v.
s.c.

C)

Test Item		Anti-PSMA-Ama1		Anti-CD37-Ama1		Anti-PSMA-Ama2	
Route		**s.c.**	i.v.	**s.c.**	i.v.	**s.c.**	i.v.
Half-Life	day	**12.5**	11.4	**13.1**	8.6	**13.8**	11.2
Cmax_D	kg·µg/mL/mg	**13.0**	16.9	**12.2**	16.5	**7.54**	18.2
AUCINF_D_obs	day·kg·ug/mL/mg	**210**	189	**201**	120	**161**	147

D)

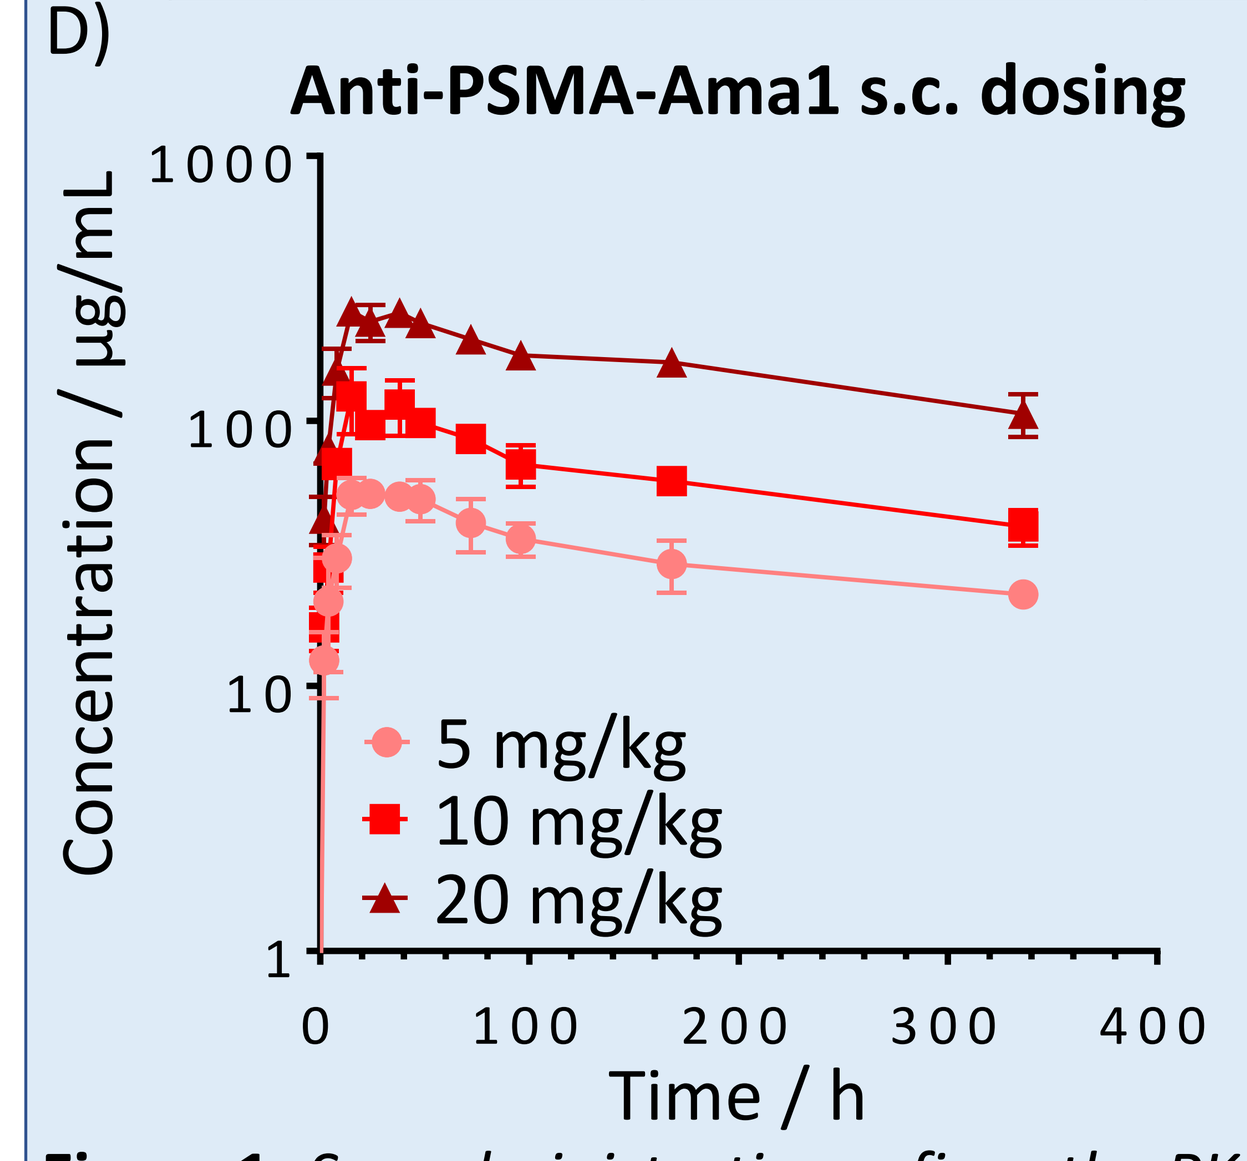
Anti-PSMA-Ama1 s.c. dosing
Concentration / µg/mL
Time / h
5 mg/kg
10 mg/kg
20 mg/kg

E)

Test Item	Route	MTD [mg/kg]
Anti-PSMA-Ama2	i.v.	6
	s.c.	**20**
Anti-PSMA-Ama3	i.v.	30
	s.c.	**40**
Anti-DIG-Ama2	i.v.	10
	s.c.	**15**

Figure 1: *S.c. administration refines the PK profile of ATACs and improves the tolerability in mice*
A) Experimental study design of a PK study in CB17-SCID mice after s.c. or i.v. single dose ATAC administration
B) Concentration of Anti-PSMA-Ama1 (left) or Anti-CD37-Ama1 (right) in serum over time after s.c. or i.v. administration
C) Serum PK parameters after s.c. or i.v. administration of ATACs in mice
D) PK profile of Anti-PSMA-Ama1 injected s.c. at three different doses
E) MTDs of ATACs after single i.v. or s.c. administration in CB17-SCID mice

S.c. administration changed the PK profile of ATACs towards longer half-lives, reduced Cmax and slightly increased AUC as compared to i.v. treatment. This effect was independent of the antibody (Anti-CD37 or anti-PSMA) and the amatoxin (Ama1 or Ama2) and which showed dose linearity.
As previous experiments have shown that the tolerability of ATAC®-based ADCs is primarily determined by the Cmax levels, the impact of the application route on the tolerability of a single dose ATAC was tested in CB17-SCID mice.
S.c. administration improved the MTD of a single dose of ATAC®-based ADCs by 30% to 300% as compared to i.v. treatment independent of the antibody (Anti-PSMA or non-targeting Anti-DIG) and the amatoxin (Ama2 or Ama3) used as payload (Figure 1 E).

2. The potent anti-tumor efficacy of ATAC®-based ADCs is maintained after s.c. administration

The effect of s.c. dosing on the anti-tumor efficacy of ATAC®-based ADCs was investigated in human cancer cell line xenograft models as previous results suggested that the efficacy of ATACs is driven by serum AUC (Figure 2).

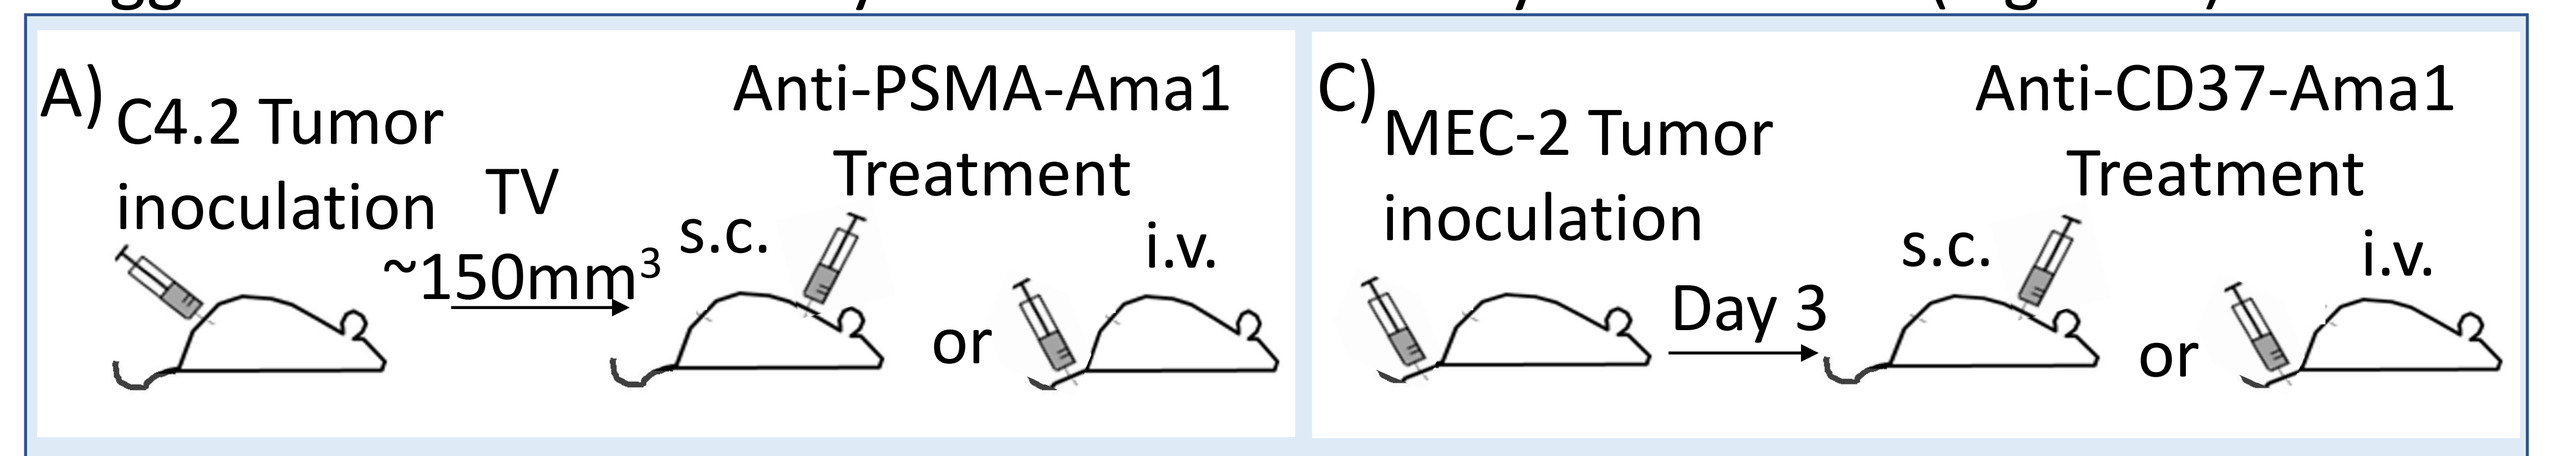
A) C4.2 Tumor inoculation
TV ~150mm3
Anti-PSMA-Ama1 Treatment
s.c.
i.v.
or
C) MEC-2 Tumor inoculation
Day 3
Anti-CD37-Ama1 Treatment
s.c.
i.v.
or

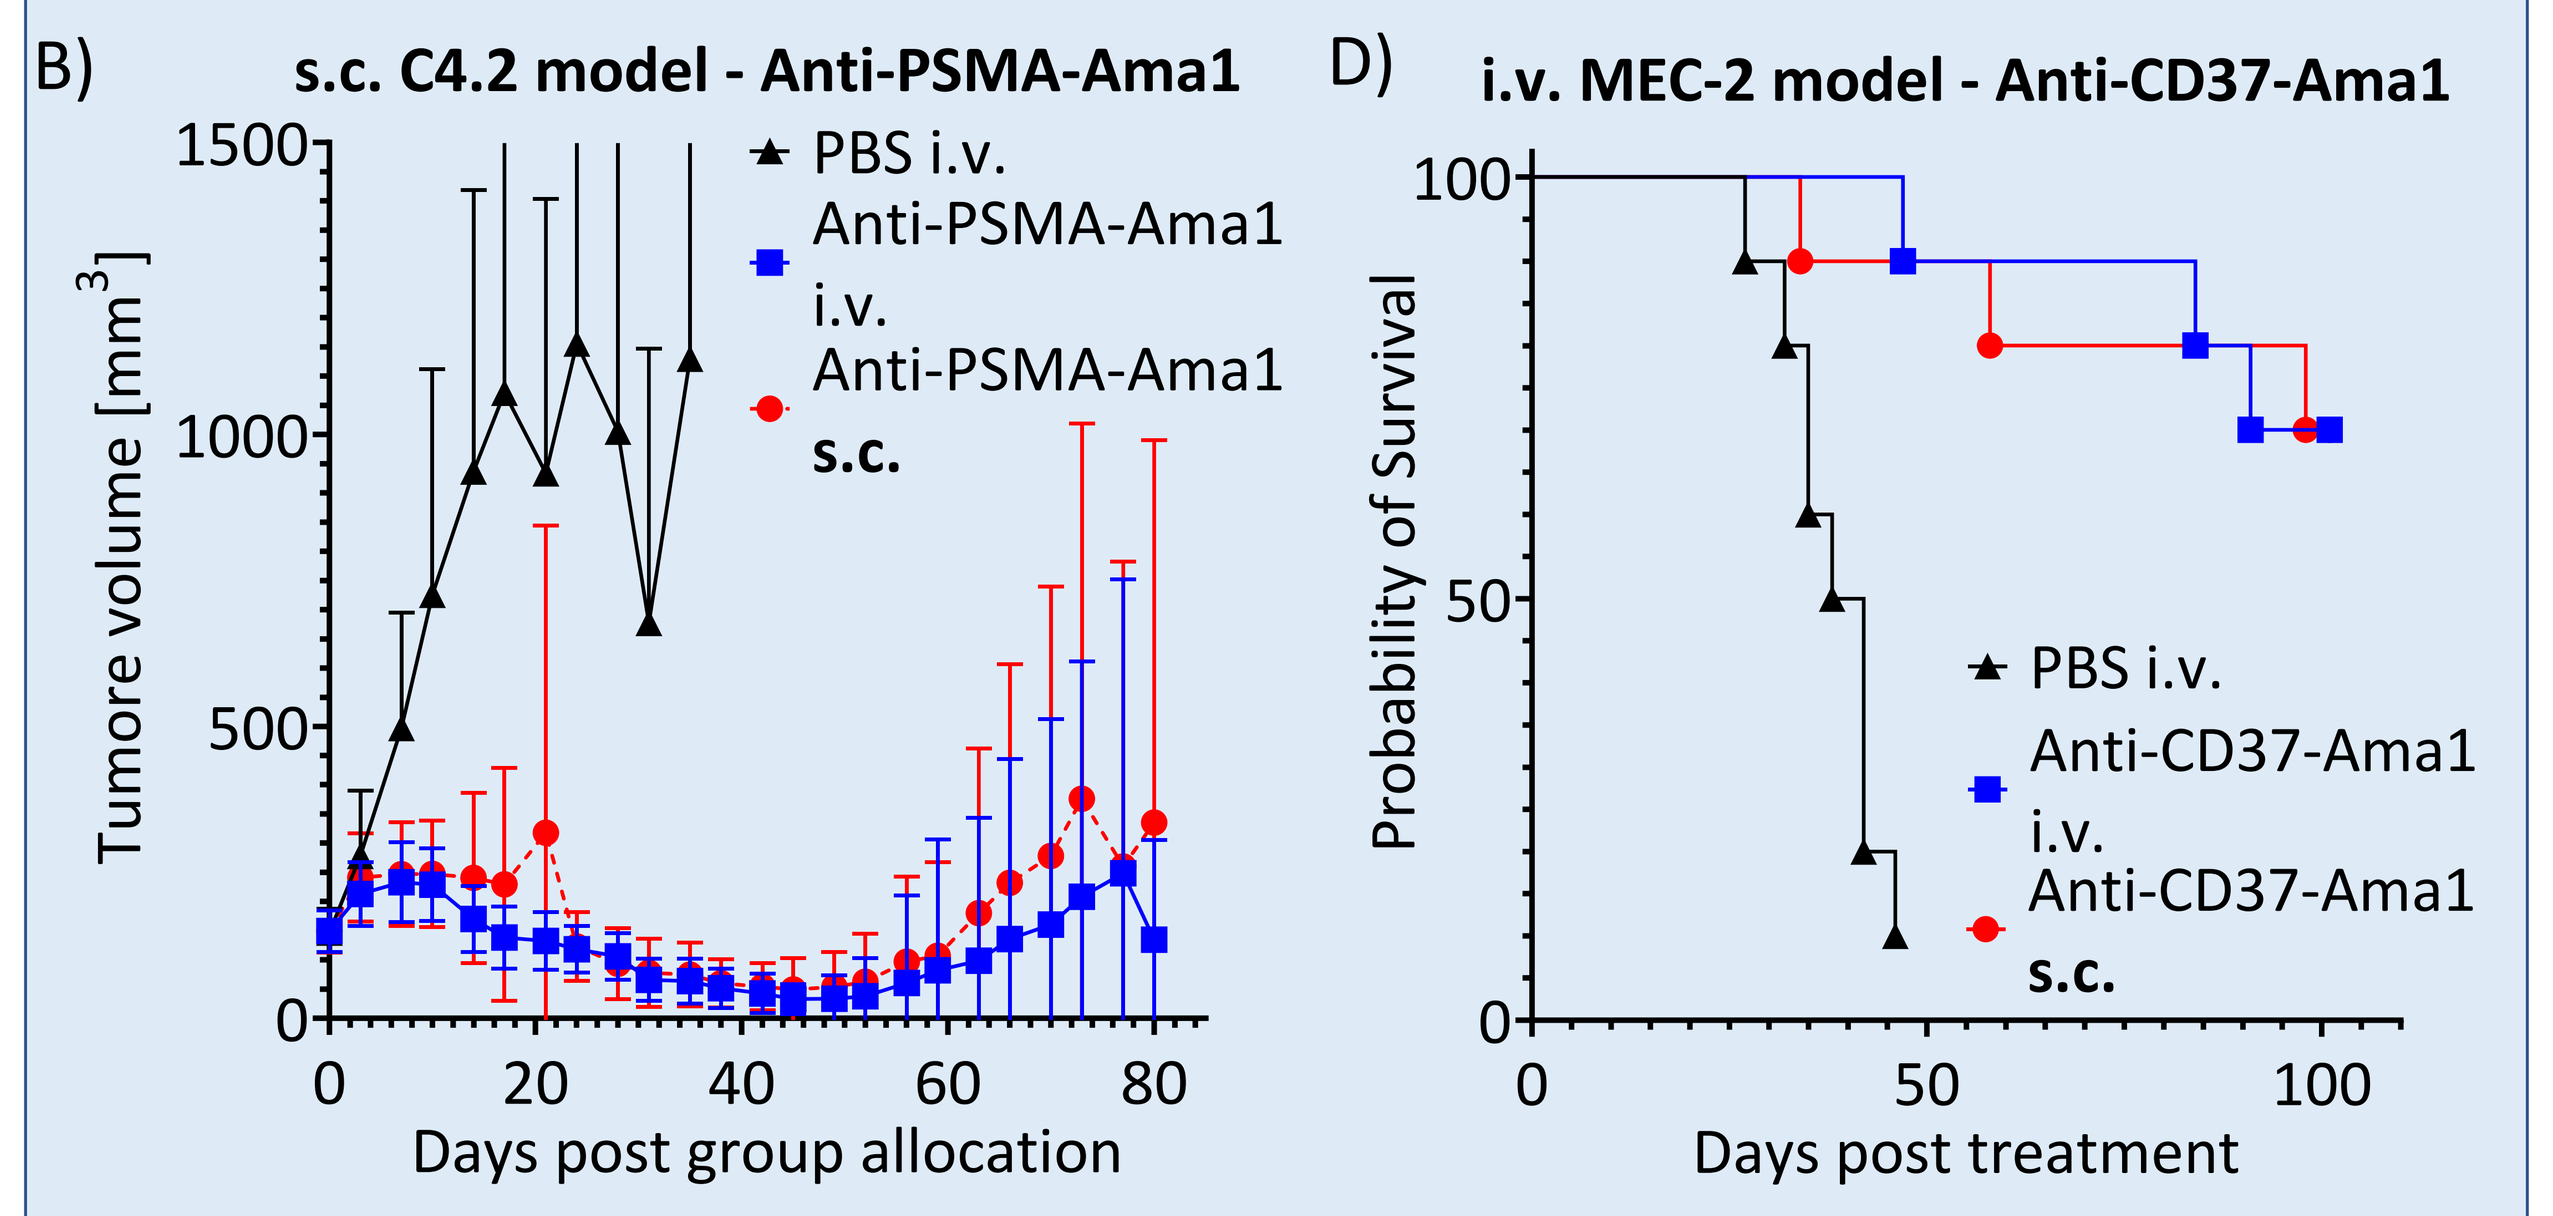
B) s.c. C4.2 model - Anti-PSMA-Ama1
Tumore volume [mm3]
Days post group allocation
PBS i.v.
Anti-PSMA-Ama1 i.v.
Anti-PSMA-Ama1 s.c.
D) i.v. MEC-2 model - Anti-CD37-Ama1
Probability of Survival
Days post treatment
PBS i.v.
Anti-CD37-Ama1 i.v.
Anti-CD37-Ama1 s.c.

Figure 2: *The application route does not impact the anti-tumor efficacy of ATAC®-based ADCs*
A) and C) Experimental set-up of an s.c. human prostate C4.2 xenograft model (A) or an i.v. human B-cell leukemia MEC-2 xenograft model (C) treated with a single i.v. or s.c. dose of Anti-PSMA-Am1 or Anti-CD37-Ama1, respectively.
B) Volume of s.c. C4.2 tumors either treated with the same single dose of Anti-PSMA-Ama1 i.v. or s.c. on day 1. Mean and SD of 10 animals per group is shown.
D) Survival plot of mice inoculated i.v. with MEC-2 cells on day 0 and treated with the same single i.v. or s.c. dose of Anti-CD37-Ama1 on day 3.

A s.c. dose of Anti-PSMA-Ama1 led to a strong growth reduction of C4.2 tumors as compared to the control group that was comparable to the effect caused by the same dose given as i.v. injection. Similar results were also obtained with a single dose of Anti-CD37-Ama1 in an i.v. MEC-2 xenograft model.
These data demonstrate that s.c. dosing maintains the potent anti-tumor effect known for ATAC®-based ADCs.

3. S.c. administration of ATAC®-based ADCs leads to a changed PK profile and improved tolerability in Cynomolgus monkeys

Having demonstrated that s.c. dosing refines the PK profile and increases the tolerability in mice, it was investigated if a similar effect is also observed in Cynomolgus monkeys, the relevant tox species for ATACs. Therefore, a single 7.5 mg/kg s.c. or i.v. dose of Anti-PSMA-Ama1, which is drug candidate HDP-103, was given on day 1. If tolerated, the dose was increased to 10 mg/kg on day 22 and the PK profile was measured (Figure 3).

A)

Test Item		Anti-PSMA-Ama1 (HDP-103)	
Dose	mg/kg	7.5	7.5
Route		i.v.	**s.c.**
Half-Life	day	8.25	**9.5**
Cmax_D	kg·µg/mL/mg	29.2	**12.8**
AUCINF_D_obs	day·kg·ug/mL/mg	161.5	**216**

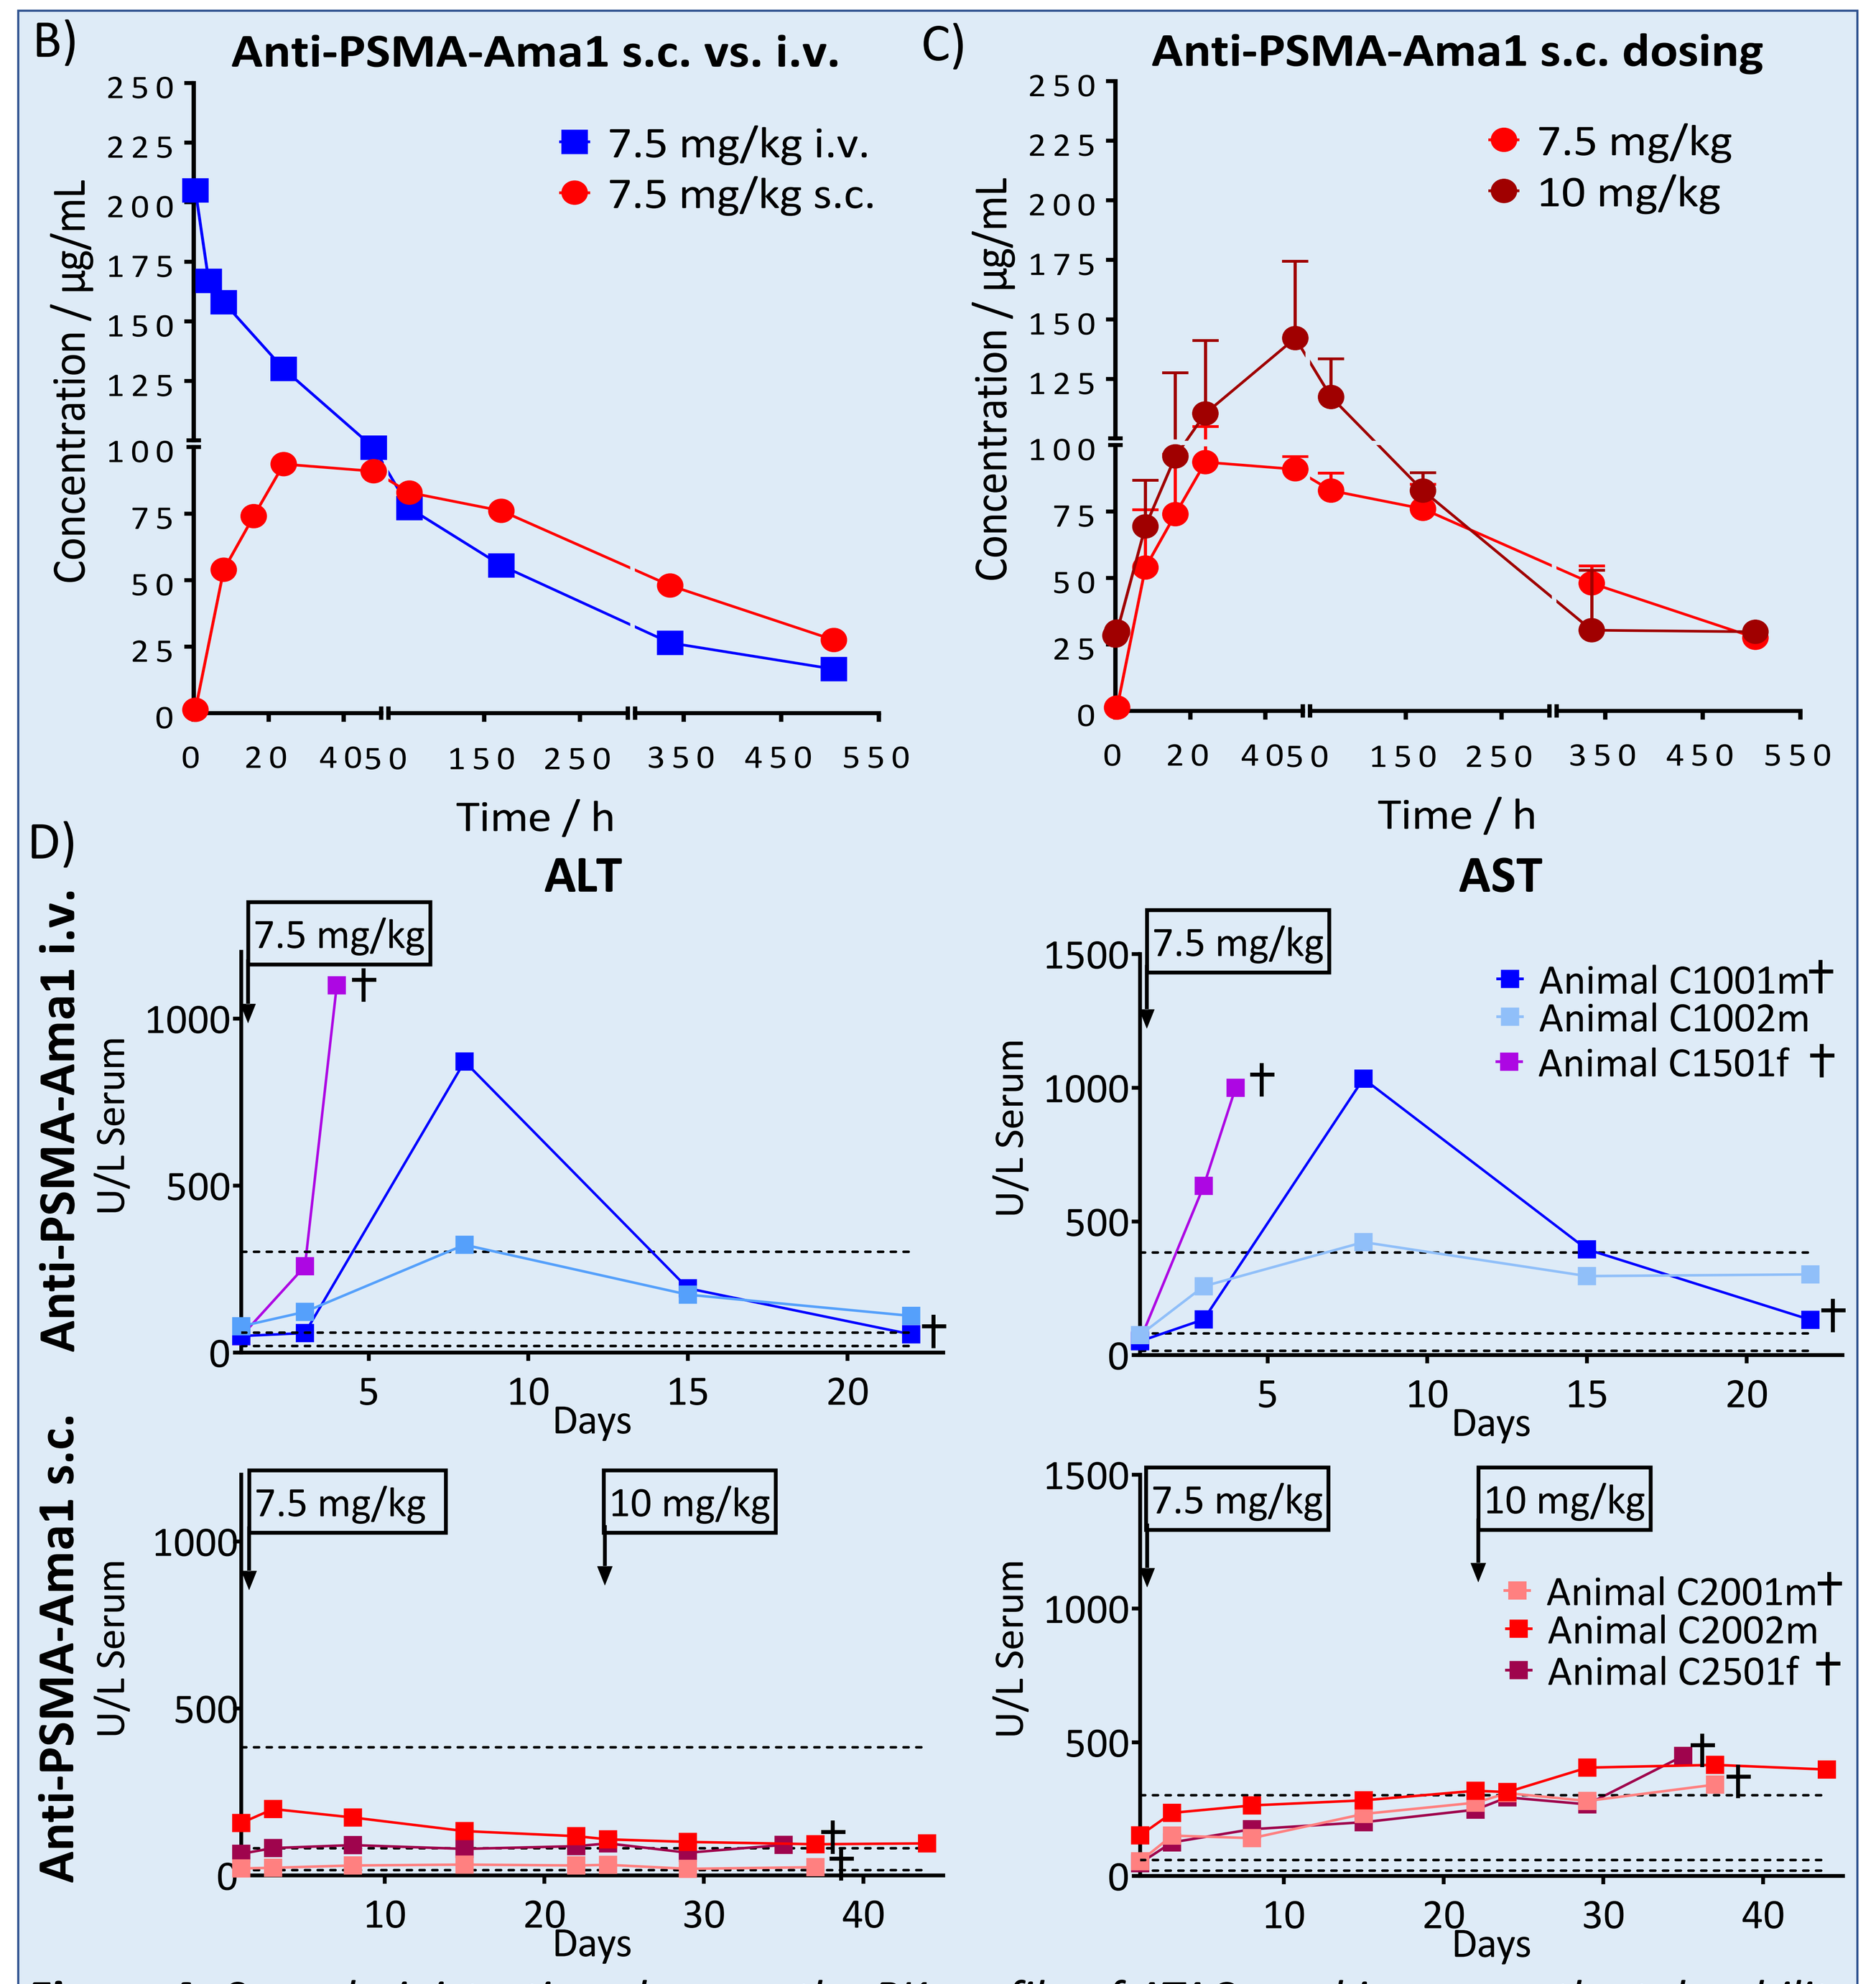
B) Anti-PSMA-Ama1 s.c. vs. i.v.
7.5 mg/kg i.v.
7.5 mg/kg s.c.
Concentration / µg/mL
Time / h
C) Anti-PSMA-Ama1 s.c. dosing
7.5 mg/kg
10 mg/kg
D) ALT
AST
Anti-PSMA-Ama1 i.v.
Anti-PSMA-Ama1 s.c.
U/L Serum
Days
7.5 mg/kg
10 mg/kg
Animal C1001m†
Animal C1002m
Animal C1501f †
Animal C2001m†
Animal C2002m
Animal C2501f †

Figure 4: *S.c. administration changes the PK profile of ATACs and improves the tolerability in Cynomolgus monkeys*
A) Serum PK parameters after a single s.c. or i.v. dose of Anti-PSMA-Ama1 in monkeys
B) Concentration of Anti-PSMA-Ama1 in serum over time after s.c. or i.v. administration (only animals that tolerated the dose were considered)
C) PK profile of Anti-PSMA-Ama1 injected s.c. at two different doses
D) ALT and AST levels in monkey serum after s.c. or i.v. treatment with Anti-PSMA-Ama1

The changes in the PK profile that were observed with s.c. dosing in mice were confirmed in monkey:. A longer half-life, a reduced Cmax and a slightly increased AUC were achieved by s.c. as compared to i.v. treatment.
This effect also translated into reduced liver toxicity and improved tolerability of HDP-103 after s.c. dosing in monkeys. The MTD of HDP-103 in Cynomolgus monkeys was improved from 5 to 7.5 mg/kg by s.c. dosing.

4. S.c. administration improves the therapeutic index of the ATAC®-based ADC Anti-PSMA-Ama1 (HDP-103)

The therapeutic index (TI) is a comparison between the tolerated dose of a drug and the dose required for efficacy and is thus a pre-clinical measurement for relative drug safety.
S.c. administration increased the TI of HDP-103 by 50% from an TI of 8 after i.v. administration to a TI of 12 after s.c. administration (Figure 5).

A)

$$TI = \frac{MTD_{monkey}}{MED_{mouse}converted}$$

B)

Conjugate	Route	TI
Anti-PSMA-Ama1 (HDP-103)	i.v.	8
	s.c.	**12**

Figure 5: *S.c. administration increases the TI of HDP-103*
A) Formula for the calculation of the TI with MTD_{monkey} = the maximal tolerated dose in Cynomolgus monkey and MED_{mouse}converted = minimal effective dose (= dose that leads to TV reduction below the start value for at least 1 week) in mouse models converted to monkey by the body surface area (/4)
B) TI of HDP-103 after i.v. or s.c. administration

Conclusion

ATAC®-based ADCs are a novel and promising class of ADCs with a new mode of action. Currently, HDP-101 is being tested in a first-in-human phase I/II clinical trial in multiple myeloma patients[5]. As long as no tolerability and efficacy data in humans are available, the therapeutic index (TI), which is based on the tolerability in Cynomolgus monkeys and the efficacy in mouse models, is the best measure to estimate the clinical success of a drug.
Parameters that can influence the TI are the drug structure itself but also the treatment schedule and administration route. Currently, ADCs and ATACs are administered via the intravenous route. As the administration route has an impact on the PK profile of the drug, the aim of the study was to investigate how s.c. instead of i.v. dosing influences PK, tolerability and efficacy of ATAC®-based ADCs. It was demonstrated that s.c. dosing leads to an increased half-life and reduced serum Cmax values of ATACs in mice and monkeys as compared to i.v. dosing independent of antibody and amatoxin variant. These changes were translated into an improved tolerability in both species. At the same time the anti-tumor efficacy of ATACs in human cell line derived cancer models in mice is comparable after s.c. or i.v. dosing. The improved tolerability in combination with the maintained efficacy leads to an improved TI of HDP-103.
The present study demonstrates that s.c. dosing not only refines the pharmacokinetic distribution of ATACs but may improve the TI in patients. Thus, s.c. dosing may represent a promising route of administration for ATAC®-based ADCs also in humans.

References

(1) Pahl A *et al.* in Cytotoxic Payloads for Antibody–Drug Conjugates Chapter 19; The Royal Society of Chemistry 2019
(2) Li Y. *et al.* J Sci Transl Med. 2021; 13(580):eabc6894
(3) Figueroa-Vazquez V *et al.;* Mol Cancer Ther 2021; 20:367–78
(4) Kim J, De Jesus O; StatPearls [Internet]. Treasure Island (FL); 2022
(5) Clinical study NCT04879043

ATAC® is a registered EU trademark of Heidelberg Pharma Research GmbH , No 017988594

Contact

Heidelberg Pharma Research GmbH
Gregor-Mendel-Straße 22
68526 Ladenburg
Germany

Phone: +49-6203-1009 0
Email: info@hdpharma.com
https://www.heidelberg-pharma.com